IDWEEK 2020 | Poster number: 1589

Activity of Meropenem-Vaborbactam and Comparators Against Globally Disseminated *Klebsiella pneumoniae* Sequence Type 258

Dee Shortridge, Lalitagauri Deshpande, Tim Doyle, Jennifer Streit, Mariana Castanheira
JMI Laboratories, North Liberty, IA, USA

Introduction

- Meropenem-vaborbactam (MVB) is a combination of a carbapenem and a β-lactamase inhibitor active against β-lactamases, including serine carbapenemases (Castanheira, Huband et al. 2017).
- MVB recently was approved in the US and Europe for the treatment of complicated UTIs, including acute pyelonephritis, and is approved in Europe for treatment of complicated intra-abdominal infections, hospital-acquired bacterial pneumonia, ventilator-associated pneumonia, and bacteremia (Melinta Therapeutics 2019).
- Carbapenemase-producing *Enterobacterales*, particularly *Klebsiella pneumoniae,* have spread worldwide and are considered endemic in various countries (Castanheira, Deshpande et al. 2019).
- Globally, 60–70% of carbapenemase-producing *K. pneumoniae* belong to Clonal Group 258, of which sequence type (ST) 258 and ST11 are the most common members (Rojas, Weinstock et al. 2017).
- Multiple outbreaks have been associated with bla_{KPC}-expressing ST258 (Chen, Mathema et al. 2014).
- In this study, we examined the susceptibilities of 130 ST258 isolates collected as a part of the SENTRY global surveillance program.

Materials and Methods

- As a part of the SENTRY Antimicrobial Surveillance Program for meropenem-vaborbactam from 2016–2019, 10,671 *K. pneumoniae* were collected.
- Isolates were tested for susceptibility against meropenem-vaborbactam and comparator agents using the reference broth microdilution method as described by the Clinical and Laboratory Standards Institute M07 (2018) and M100 (2020) documents.
 - Vaborbactam was tested at a fixed concentration of 8 mg/L.
 - Quality control (QC) was performed according to the CLSI M100 (2020) criteria.
 - All QC MIC results were within acceptable ranges.
 - Categorical interpretations for all comparator agents were those criteria found in the European Committee on Antimicrobial Susceptibility Testing (EUCAST) breakpoint tables (2020), the CLSI M100 (2020), or the US Food and Drug Administration (FDA) website (2020).
 - Multidrug-resistant (MDR) isolates were identified as nonsusceptible to at least 1 agent in 3 or more antimicrobial classes.
 - Extensively drug-resistant (XDR) isolates were identified as nonsusceptible to 1 or more agents in all but 1 or 2 antimicrobial classes.
- Carbapenem-resistant (CR) *K. pneumoniae* was defined as any isolate exhibiting doripenem, imipenem, and/or meropenem MIC values at ≥2 mg/L.
 - CR isolates were submitted to whole genome sequencing on a MiSeq (Illumina, San Diego, California, USA) instrument targeting a 30X coverage.
 - Sequences were *de novo* assembled, searched for the presence of acquired carbapenemases using a curated library, and applied the criteria of >94% sequencing identity and 40% minimum length coverage.
 - The sequence type was determined by whole genome sequencing analysis.

Results

- The most common infection type from which *K. pneumoniae* was isolated was bloodstream infection (n=3,346), followed by pneumonia in hospitalized patients (n=2,881).
 - 1,277 isolates were CR (12.0%).
- A subset of 130 *K. pneumoniae* ST258 isolates were selected for further study. Susceptibilities of MVB and comparators for these isolates are shown in Table 1.
 - The MIC distributions for the isolates and comparators are shown in Table 2.
- MVB inhibited 99.2% of the isolates and was the most active agent overall; only 23.1% of these isolates were meropenem susceptible.
- Out of 130 isolates, 127 isolates were MDR and 89 isolates were XDR.
 - Tigecycline was the most active comparator with 98.5% susceptible.
 - Amikacin had 30.8% susceptible.
- The geographic distribution of ST258 isolates is shown in Figure 1.
 - The US had the most ST258 isolates (56, 43.1%), followed by Greece (32, 24.6%) and Brazil (22, 16.9%).
- The distribution of the 98 carbapenemase-producing ST258 isolates is shown in Figure 2.
 - KPC-2 was the most common carbapenemase; 73 isolates produced KPC-2 (including 2 KPC-2-like).
 - The US had 24 KPC-2 isolates, followed by Brazil and Greece, which had 17 each.
 - KPC-3 was produced by 25 isolates, 19 of which were from the US.
 - One KPC-12-producing isolate was found in Greece.
 - The single NDM-1-producing isolate was from Argentina.

Conclusions

- Isolates from the internationally disseminated *K. pneumoniae* clone ST258 were found in 6 countries.
 - ST258 was most frequently found in the US (43.1%).
 - KPC-2 was the most common carbapenemase among CR isolates.
 - Only 1 non-KPC carbapenemase-producing isolate was found, an NDM-1 in Argentina.
- MVB had potent activity against ST258 isolates, including those isolates producing KPC.
- MVB may be useful for the treatment of infections caused by MDR and XDR *K. pneumoniae*.

Table 1 Susceptibilities of meropenem-vaborbactam and comparators to *K. pneumoniae* ST258 isolates by country

Organisms by country of origin (n)	% susceptible using CLSI/FDA breakpoints[a]					
	Meropenem-vaborbactam	Meropenem	Amikacin	Piperacillin-tazobactam	Tigecycline	Colistin
All (130)	99.2	23.1	30.8	6.9	98.5	67.4
US (56)	100.0	25.0	41.1	8.9	98.2	81.8
Greece (32)	100.0	28.1	18.8	12.5	100.0	62.5
Brazil (22)	100.0	18.2	27.3	0.0	100.0	36.4
Argentina [b] (15)	93.3	20.0	20.0	0.0	93.3	73.3
Italy (3)	100.0	0.0	0.0	0.0	100.0	33.7
Romania (2)	100.0	0.0	100.0	0.0	100.0	0.0

[a] CLSI M100 (2020). FDA breakpoints are shown for tigecycline. For colistin, %intermediate is shown; no susceptible category is defined by CLSI.
[b] 1 isolate producing NDM-1.

Table 2 MIC distributions of meropenem-vaborbactam for 130 *K. pneumoniae* ST258 isolates

Antimicrobial Agent	Dilution (mg/L)														Total	MIC_{50}	MIC_{90}
	0.015	0.03	0.06	0.12	0.25	0.5	1	2	4	8	16	32	64	>			
Meropenem-vaborbactam	24	40	9	8	14	12	15	7	0	0	0	0		1	130	0.06	1
	18.5%	49.2%	56.2%	62.3%	73.1%	82.3%	93.8%	99.2%	99.2%	99.2%	99.2%	99.2%		100.0%			
Meropenem	7	9	5	3	1	2	3	1	7	7	19	18		48	130	32	>32
	5.4%	12.3%	16.2%	18.5%	19.2%	20.8%	23.1%	23.8%	29.2%	34.6%	49.2%	63.1%		100.0%			
Imipenem				25	1	3	0	1	7	9				84	130	>8	>8
				19.2%	20.0%	22.3%	22.3%	23.1%	28.5%	35.4%				100.0%			
Tigecycline				0	8	62	38	20	2						130	0.5	2
				0.0%	6.2%	53.8%	83.1%	98.5%	100.0%								
Amikacin					0	4	4	2	6	4	20	49		41	130	32	>32
					0.0%	3.1%	6.2%	7.7%	12.3%	15.4%	30.8%	68.5%		100.0%			
Colistin[a]			3	50	29	2	2	1	3	8				31	129	0.25	>8
			2.3%	41.1%	63.6%	65.1%	66.7%	67.4%	69.8%	76.0%				100.0%			
Piperacillin-tazobactam							0	1	2	4	2	2	1	118	130	>64	>64
							0.0%	0.8%	2.3%	5.4%	6.9%	8.5%	9.2%	100.0%			

CLSI (2020), FDA for tigecycline
Breakpoints are indicated with green for susceptible, yellow for intermediate, and orange for resistant.
[a]Colistin has no susceptible CLSI breakpoint.

Figure 1 Geographic distribution of ST258 in the SENTRY Surveillance Program

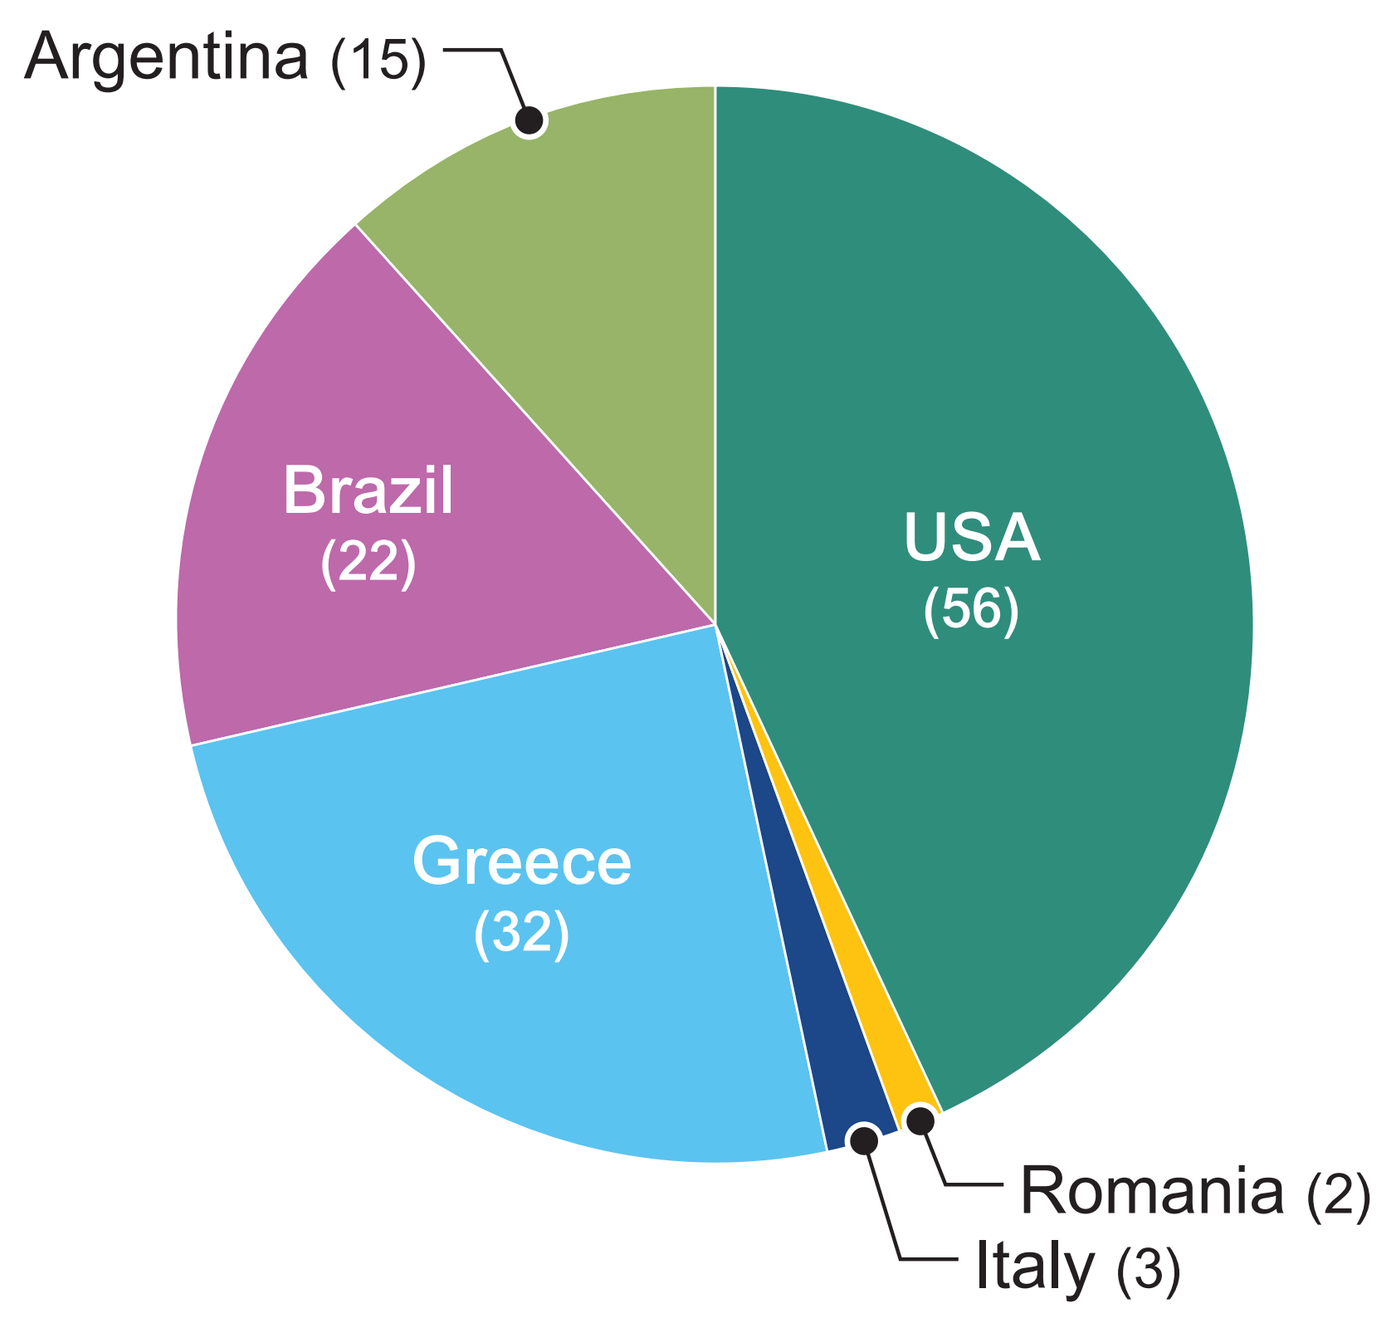
Argentina (15)
Brazil (22)
USA (56)
Greece (32)
Romania (2)
Italy (3)

Figure 2 Geographic distribution of ST258 isolates containing carbapenemase (SENTRY)

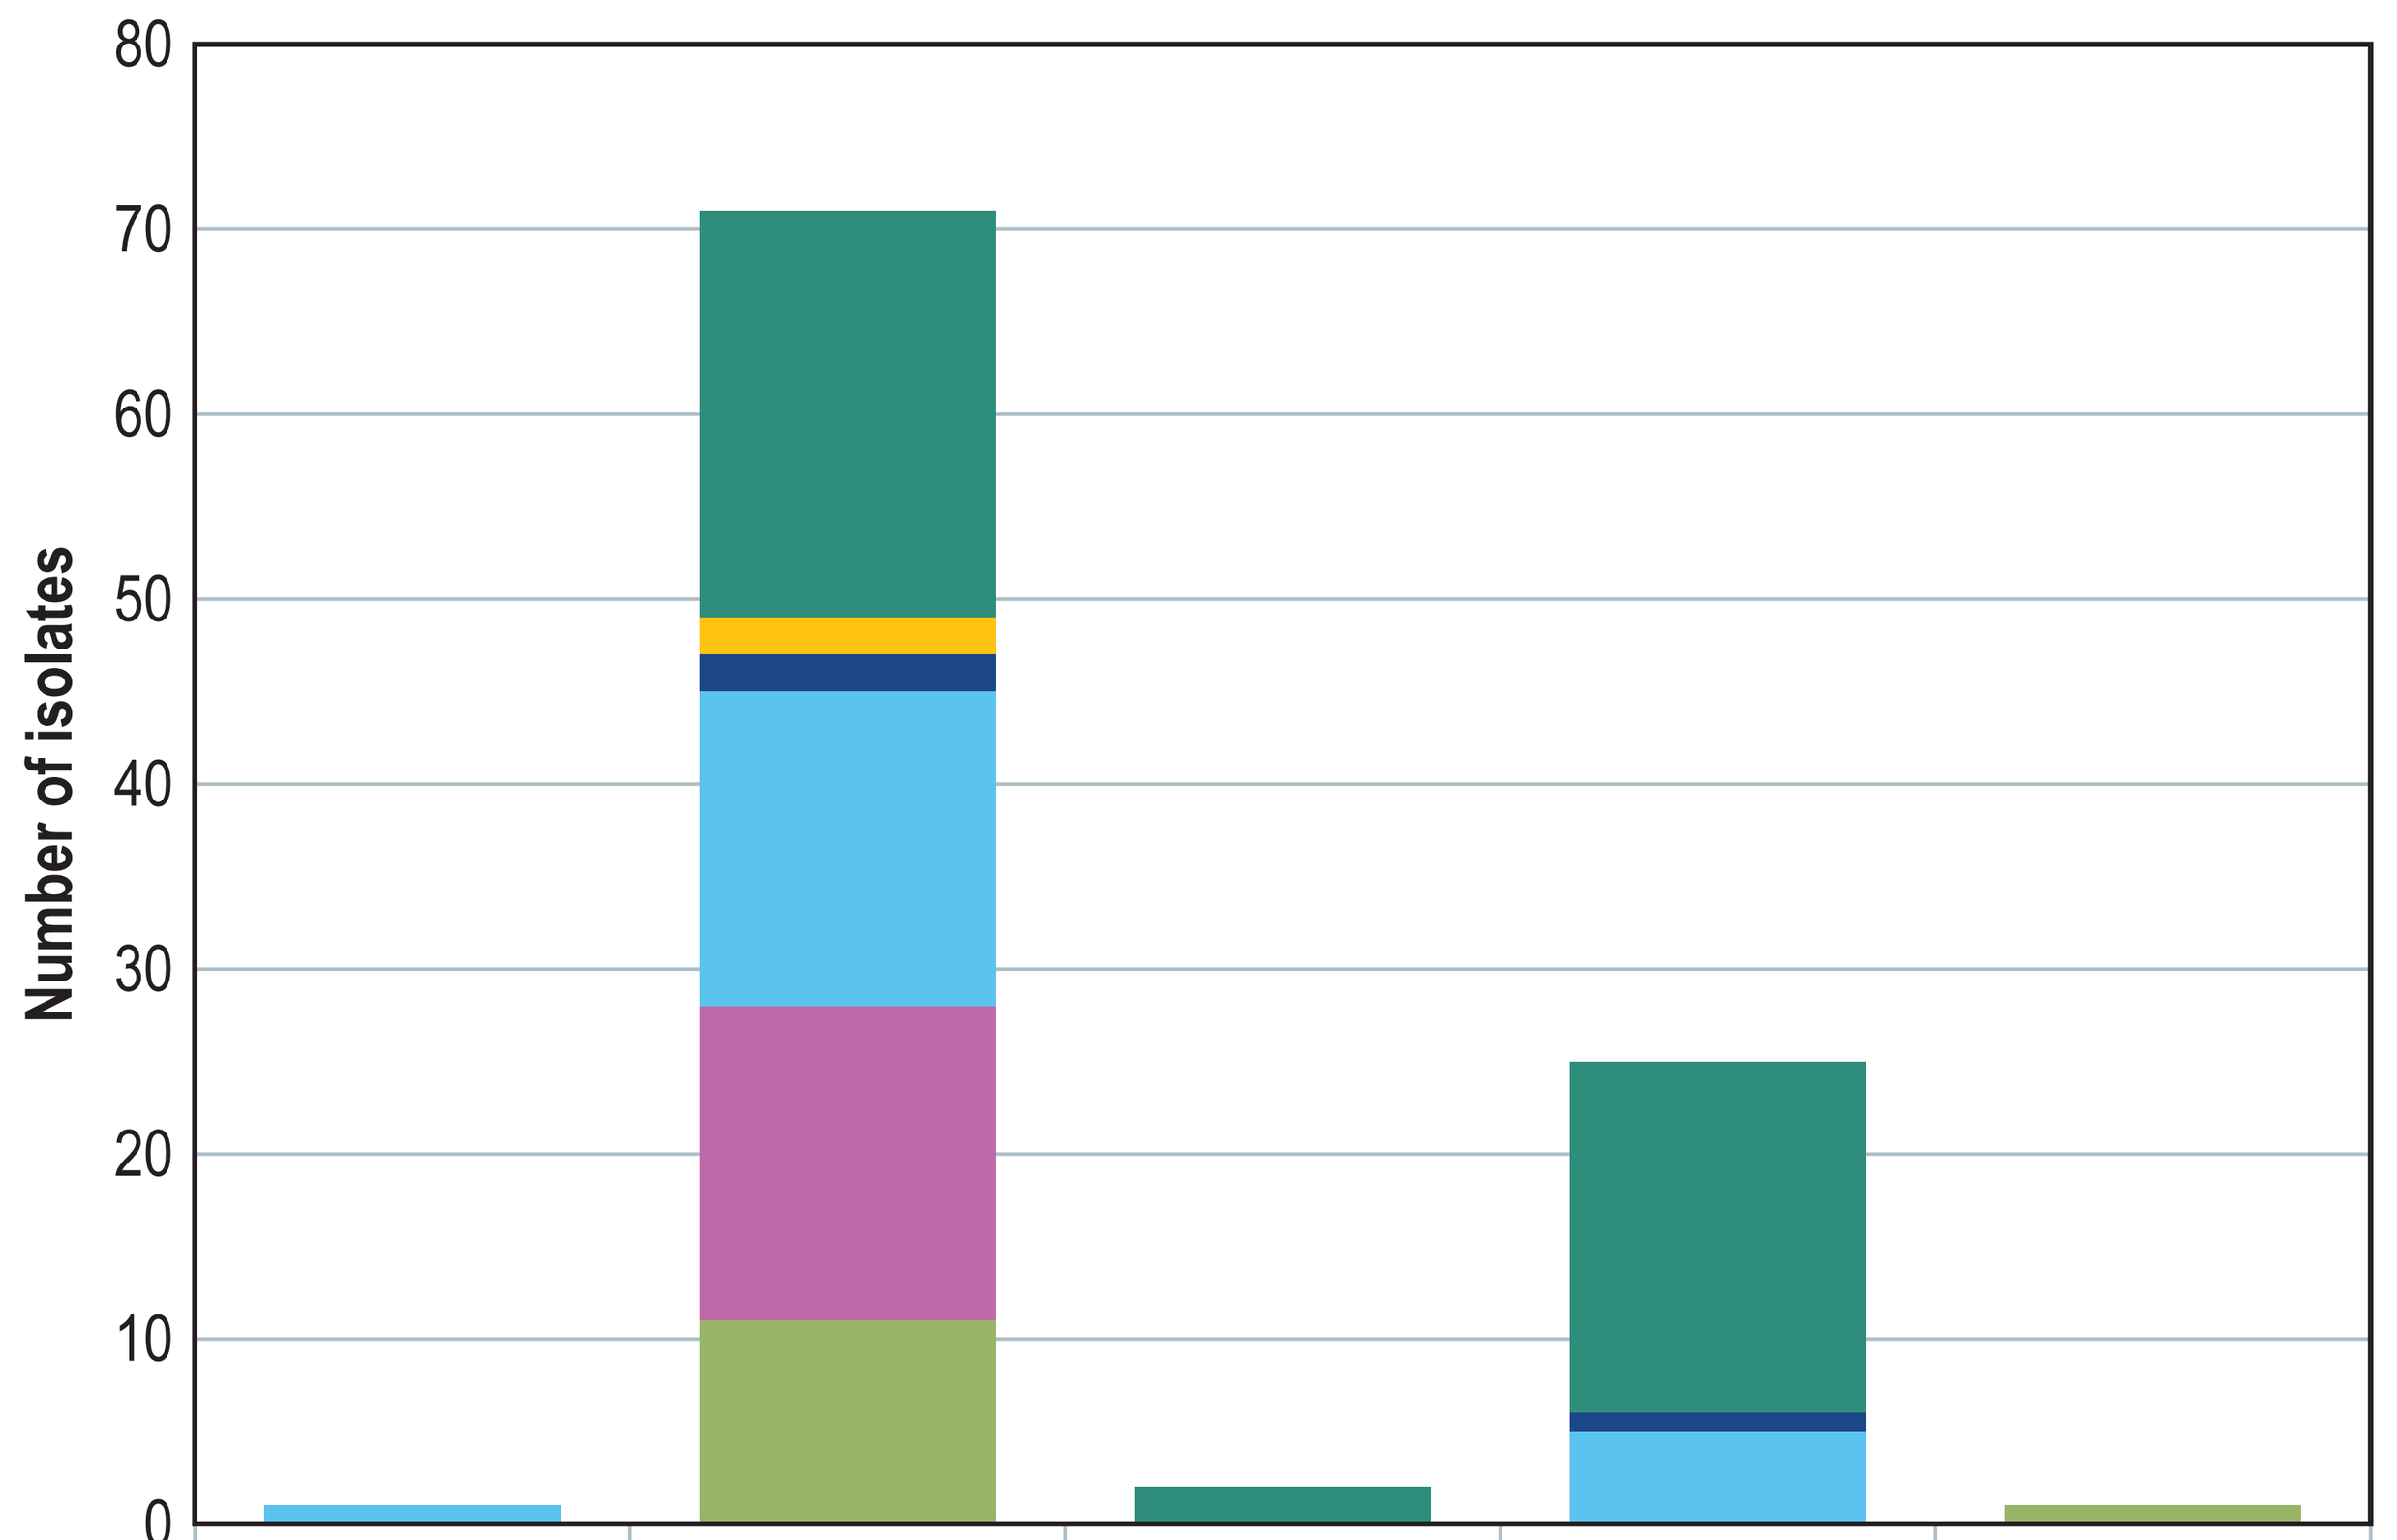
Number of isolates
0
10
20
30
40
50
60
70
80

	KPC-12	KPC-2	KPC-2-like	KPC-3	NDM-1
USA		22	2	19	
Romania		2			
Italy		2		1	
Greece	1	17		5	
Brazil		17			
Argentina		11			1

Acknowledgements

This study was supported by Melinta Therapeutics, Inc.

References

Castanheira, M., L. M. Deshpande, R. E. Mendes, R. Canton, H. S. Sader and R. N. Jones (2019). “Variations in the Occurrence of Resistance Phenotypes and Carbapenemase Genes Among Enterobacteriaceae Isolates in 20 Years of the SENTRY Antimicrobial Surveillance Program.” *Open Forum Infect Dis* 6(Suppl 1): S23–S33.

Castanheira, M., M. D. Huband, R. E. Mendes and R. K. Flamm (2017). “Meropenem-Vaborbactam Tested against Contemporary Gram-Negative Isolates Collected Worldwide during 2014, Including Carbapenem-Resistant, KPC-Producing, Multidrug-Resistant, and Extensively Drug-Resistant Enterobacteriaceae.” *Antimicrob Agents Chemother* 61(9).

Chen, L., B. Mathema, K. D. Chavda, F. R. DeLeo, R. A. Bonomo and B. N. Kreiswirth (2014). “Carbapenemase-producing Klebsiella pneumoniae: molecular and genetic decoding.” *Trends Microbiol* 22(12): 686–696.

CLSI (2018). M07 Eleventh edition. Methods for Dilution Antimicrobial Susceptibility Testing for Bacteria That Grow Aerobically. Wayne, PA, Clinical and Laboratory Standards Institute.

CLSI (2020). M100 30th edition. Performance Standards for Antimicrobial Susceptibility Testing. Wayne, PA, Clinical and Laboratory Standards Institute.

EUCAST (2020). “The European Committee on Antimicrobial Susceptibility Testing. Breakpoint tables for interpretation of MICs and zone diameters.”

FDA (2020). “FDA-Recognized Antimicrobial Susceptibility Test Interpretive Criteria.” Retrieved August 17, 2020, from https://www.fda.gov/drugs/development-resources/fda-recognized-antimicrobial-susceptibility-test-interpretive-criteria.

Melinta Therapeutics, Inc. (2019). Vabomere Prescribing Information. *FDA*.

Rojas, L. J., G. M. Weinstock, E. De La Cadena, L. Diaz, R. Rios, B. M. Hanson, J. S. Brown, P. Vats, D. S. Phillips, H. Nguyen, K. M. Hujer, A. Correa, M. D. Adams, F. Perez, E. Sodergren, A. Narechania, P. J. Planet, M. V. Villegas, R. A. Bonomo and C. A. Arias (2017). “An Analysis of the Epidemic of Klebsiella pneumoniae Carbapenemase-Producing K. pneumoniae: Convergence of Two Evolutionary Mechanisms Creates the “Perfect Storm”.” *J Infect Dis* 217(1): 82–92.

Contact

Dee Shortridge, PhD
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, Iowa 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: dee-shortridge@jmilabs.com