1867

Meropenem-Vaborbactam (VABOMERE) vs. Best Available Therapy for Carbapenem-Resistant Enterobacteriaceae Infections in TANGO II: Primary Outcomes by Site of Infection

The Medicines Company
3013 Science Park Rd.
San Diego, CA 92121 USA
8 Sylvan Way
Parsippany, NJ 07054 USA

Richard Wunderink[1], Evangelos J. Giamarellos-Bourboulis[2], Galia Rahav[3], Amy Mathers[4], Matteo Bassetti[5], Joseph Solomkin[6], Elizabeth Alexander[7], Jeff Loutit[8], Shu Zhang[7], Michael Dudley[8], Keith S. Kaye[9]

[1]Northwestern University Feinberg School of Medicine, Chicago, IL, USA; [2]National and Kapodistrian University of Athens, Medical School, Athens, Greece; [3]Sheba Medical Center, Tel Hahsomer, Israel; [4]University of Virginia Health System, Charlottesville, VA, USA; [5]University of Udine, Udine, Italy; [6]University of Cincinnati College of Medicine, Cincinnati, OH, USA; [7]The Medicines Company, Parsippany, NJ, USA; [8]The Medicines Company, San Diego, CA, USA; [9]University of Michigan Medical School, Ann Arbor, MI, USA

Abstract

Introduction
Meropenem-vaborbactam (M-V) is a beta-lactamase inhibitor combination active against *Klebsiella pneumoniae* carbapenemase (KPC)-producing CRE. Few clinical trials of new agents have been conducted in patients with CRE.

Methods
TANGO II is a randomized, Phase 3, open-label trial in patients with infections due to known or suspected CRE, including complicated urinary tract infection (cUTI), acute pyelonephritis (AP), HABP/VABP, bacteremia, or complicated intra-abdominal infection (cIAI). Eligible patients were randomized 2:1 to monotherapy with M-V or Best Available Therapy (BAT) for 7-14 days. BAT could include (alone or in combination): a carbapenem, aminoglycoside, polymyxin B, colistin, tigecycline or ceftazidime-avibactam (monotherapy only). Enrollment was stratified by infection type and geographic region. Endpoints differed by infection: overall success (clinical cure + microbial eradication) in cUTI/AP, 28-day all-cause mortality in HABP/VABP + bacteremia, and clinical cure in cIAI. It was not powered for inferential statistical testing; results are presented descriptively.

Results
72 patients were enrolled: 43 (59.7%) had baseline CRE and comprised the microbiologic CRE modified intent-to-treat population (mCRE-MITT, primary population). In mCRE-MITT, 20 had bacteremia, 15 had cUTI/AP, 5 had HABP/VABP, and 3 had cIAI.

Efficacy by Infection Type	M-V (N=28) n/N' (%)	BAT (N=15) n/N' (%)
Bacteremia + HABP/VABP All-Cause Mortality, Day 28	4/16 (25.0%)	4/9 (44.4%)
cUTI/AP Overall Success at End of Treatment (EOT) Overall Success at Test of Cure (TOC, EOT + 7 d)	8/11 (72.7%) 3/7 (42.9%)*	2/4 (50%) 2/4 (50%)
cIAI Clinical Cure at TOC	1/1 (100%)	0/2 (0)

AEs occurred in 84.4% of M-V patients vs. 92% on BAT. M-V was associated with fewer drug-related AEs (24.4% vs. 44%), severe AEs (13.3% vs. 28%), and serious AEs (33.3% vs. 44%) vs. BAT.

Conclusions
In this first prospective comparative trial of a beta-lactam/beta-lactamase inhibitor combination as monotherapy of CRE infections, M-V showed consistent improvement over BAT in efficacy endpoints across infections, and improved safety/tolerability. M-V appears to be an improved treatment option for CRE infections.

Methods

- Phase 3, multi-center, randomized, open-label study of adults with infections due to known or suspected CRE, including complicated urinary tract infection (cUTI), acute pyelonephritis (AP), hospital-acquired/ventilator-associated bacterial pneumonia (HABP/VABP), bacteremia, or complicated intra-abdominal infection (cIAI).
- Eligible patients were randomized 2:1 to monotherapy with M-V (2g/2g every 8h via 3-h infusion) or BAT for 7-14 days (**Figure 1**).
 - BAT included mono/combination therapy with polymyxins, carbapenems, aminoglycosides, tigecycline; or ceftazidime-avibactam alone.
- Enrollment was stratified by infection type and geographic region.
- Key inclusion criteria: known or suspected (evidence of CRE in culture or molecular testing within past 90 d) CRE pathogen, requirement of ≥7 days IV therapy, confirmed cUTI/AP, HABP/VABP, bacteremia, or cIAI.
- Key exclusion criteria: Receipt of >24 hours of potentially effective antimicrobials (unless clinical failure), immediate life-threatening disease, known infection due to NDM, VIM, IMI or OXA-encoded beta-lactamase.
- Efforts to reduce bias included onsite blinded investigator, blinded adjudication committee, and source control adjudication committee (for cIAI).

Figure 1. Study Schema

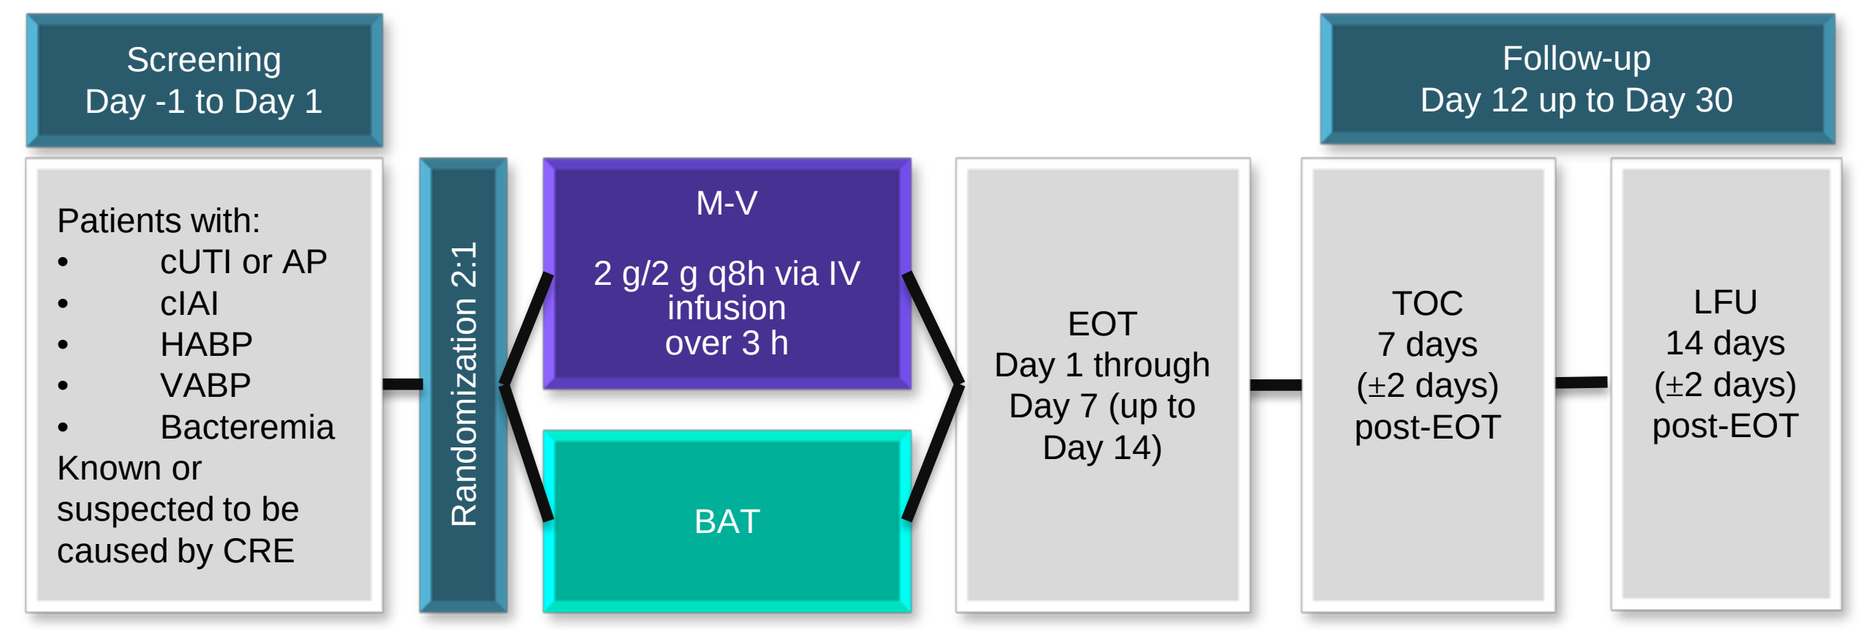
Screening
Day -1 to Day 1
Follow-up
Day 12 up to Day 30
Patients with:
• cUTI or AP
• cIAI
• HABP
• VABP
• Bacteremia
Known or suspected to be caused by CRE
Randomization 2:1
M-V
2 g/2 g q8h via IV infusion over 3 h
BAT
EOT
Day 1 through Day 7 (up to Day 14)
TOC
7 days (±2 days) post-EOT
LFU
14 days (±2 days) post-EOT

EOT, end of treatment; TOC, test of cure; LFU, last follow-up.

- Study endpoints differed by infection type based on prevailing FDA guideline recommended endpoints:
 - 28-day all-cause mortality in the combined HABP/VABP and bacteremia infections.
 - Overall success (composite endpoint of clinical cure and microbial eradication) in cUTI/AP at End of Treatment (EOT) and Test of Cure (TOC).
 - Clinical cure was defined as a complete resolution of signs/symptoms such that no further antimicrobial therapy was required.
 - Clinical cure was assessed by both an unblinded (PI) and a blinded investigator (BI) at two time points: EOT and TOC. In cases where their assessments differed, clinical cure was adjudicated by the blinded adjudication committee.
 - Clinical cure in cIAI at TOC
 - For cIAI, proper source control in all patients was assessed and adjudicated by committee.
- Safety endpoints were assessed in those who received ≥1 dose of study drug.
- Results are presented descriptively. The study was not powered for inferential statistical testing.

Background

- Increasing carbapenem resistance among Enterobacteriaceae pathogens has been recognized by the CDC and WHO as an urgent antimicrobial resistance threat.[1,2]
- In the US, the production of *Klebsiella pneumoniae* carbapenemase (KPC) enzyme is the major mechanism of carbapenemase-mediated resistance in carbapenem-resistant Enterobacteriaceae (CRE).[3]
- Despite the recent approval of an antimicrobial agent that has activity against target CRE pathogens, few clinical trials have been conducted in patients with infections due to confirmed CRE pathogens.
- Meropenem-vaborbactam (M-V; VABOMERE) is a novel beta-lactam/beta-lactamase inhibitor combination with potent activity against KPC-producing CRE.
- TANGO II is a Phase 3, multi-center, randomized, open-label comparative trial designed to evaluate efficacy and safety of M-V vs. best available therapy (BAT). It is the only clinical trial of a novel agent given as monotherapy compared against BAT in patients with serious CRE infections.
- Randomization into the comparator phase of TANGO II was stopped on July 21, 2017, in accordance with recommendations of the independent Data Safety Monitoring Board, which concluded that a risk-benefit analysis of available data no longer supported randomization of patients to BAT.
- Here we report primary outcomes by site of infection in patients with confirmed CRE infection.

Results

- Of the 72 enrolled patients, 43 (59.7%) had a baseline CRE and comprised the microbiologic CRE modified intent-to-treat population (mCRE-MITT, primary population). Baseline characteristics for the mCRE-MITT population are shown in **Table 1**.
- In mCRE-MITT, 20 had bacteremia, 15 had cUTI/AP, 5 had HABP/VABP, and 3 had cIAI.
 - Of the 15 patients with cUTI/AP, 7 were cUTI and 8 were AP.
 - 5 of the 7 cUTI patients had an indwelling urinary catheter at baseline.
 - Of the 5 patients with HABP/VABP, 1 was HABP and 4 were VABP.
 - Of the 3 patients with cIAI, one was due to a perforated viscus, one was in the setting of cholangitis/cholecystitis, and one was in the setting of intra-abdominal abscess.

Table 1. Baseline Demographic and Clinical Characteristics (mCRE-MITT)

	M-V N=28	BAT N=15	Total N=43
Age, mean (standard deviation), y	63.9 (14.0)	60.2 (13.0)	62.6 (13.6)
Age cohort, n (%)			
<65 y	14 (50.0)	9 (60.0)	23 (53.5)
≥65 y	14 (50.0)	6 (40.0)	20 (46.5)
≥75 y	6 (21.4)	3 (20.0)	9 (20.9)
Female gender, n (%)	15 (53.6)	5 (33.3)	20 (46.5)
White race, n (%)	24 (85.7)	12 (80.0)	36 (83.7)
Region, n (%)			
North America	6 (21.4)	7 (46.7)	13 (30.2)
Europe	17 (60.7)	8 (53.3)	25 (58.1)
Rest of World (Israel, Colombia, Brazil, Argentina)	5 (17.9)	0 (0)	5 (11.6)
Body mass index, mean (SD)	28.4 (9.4)	25.8 (7.6)	27.5 (8.8)
Infection type, n (%)			
Bacteremia	12 (42.9)	8 (53.3)	20 (46.5)
cUTI/AP	11 (39.3)	4 (26.7)	15 (34.9)
HABP/VABP	4 (14.3)	1 (6.7)	5 (11.6)
cIAI	1 (3.6)	2 (13.3)	3 (7.0)
Baseline pathogen[a], n (%)			
Klebsiella pneumoniae	25 (89.3)	12 (80.0)	37 (86.0)
Escherichia coli	2 (7.1)	1 (6.7)	3 (7.0)
Enterobacter cloacae complex	1 (3.6)	2 (13.3)	3 (7.0)
Proteus mirabilis	0 (0)	2 (13.3)	2 (4.7)
Serratia marcescens	1 (3.6)	1 (6.7)	2 (4.7)
Enrolled as known CRE, n (%)	20 (71.4)	14 (93.3)	34 (79.1)
Enrolled as suspected CRE, n (%)	8 (28.6)	1 (6.7)	9 (20.9)
Creatinine clearance, mL/m, n (%)			
>50	22 (78.6)	9 (60.0)	31 (72.1)
30–49	3 (10.7)	2 (13.3)	5 (11.6)
20–29	1 (3.6)	2 (13.3)	3 (7.0)
<20	1 (3.6)	0 (0)	1 (2.3)
Missing	1 (3.6)	2 (13.3)	3 (7.0)
Charlson Comorbidity Index, n (%)			
≤2	4 (14.3)	1 (6.7)	5 (11.6)
3–4	3 (10.7)	2 (13.3)	5 (11.6)
5	10 (35.7)	1 (6.7)	11 (25.6)
>6	11 (39.3)	11 (73.3)	22 (51.2)
Diabetes mellitus, n (%)	9 (32.1)	7 (46.7)	16 (37.2)
Systemic inflammatory response syndrome, n (%)	12 (42.9)	6 (40.0)	18 (41.9)
Intensive care unit admission	5 (17.8)	2 (13.3)	7 (16.3)
Immunocompromised[b], n (%)	10 (35.7)	8 (53.3)	18 (41.9)
Prior treatment failure[c], n (%)	9 (32.1)	0 (0)	9 (20.9)

[a] Baseline pathogens listed occurred in 2 or more patients.
[b] Leukemia or lymphoma (not in remission), prior solid organ/stem cell transplantation, ongoing neutropenia, or active receipt of immunosuppressive medications (including high-dose systemic steroids for ≥2 weeks).
[c] Clinical evidence of prior antimicrobial failure as ascertained by the study investigator at screening and randomization.

- BAT antibiotic regimens by infection type (mCRE-MITT) are shown in **Table 2**.
- There was no consensus BAT regimen. BAT ranged from 1 to >4 drug combinations, which included ceftazidime/avibactam, polymyxins, carbapenems, aminoglycosides, and tigecycline.
- For all infection types, the primary pathogen was *Klebsiella pneumonia*. The most common molecular mechanism of carbapenem resistance was production of KPC carbapenemase (90%).
- Meropenem MIC50s and 90s were similar across both arms. For cUTI and bacteremia, the meropenem MIC50 was 64 µg/mL. For HABP/VABP and cIAI, the MIC50s were 16 and >16 µg/mL, respectively.

Table 2. BAT Antibiotic Regimens by Infection Type (mCRE-MITT)

BAT Regimen	cUTI/AP N=4 n (%)	HABP/VABP N=1 n (%)	Bacteremia N=8 n (%)	cIAI N=2 n (%)	Total N=15* n (%)
Monotherapy	2 (50)	0 (0)	2 (25)	0 (0)	4 (26.7)
Aminoglycoside	1 (25)	0 (0)	0 (0)	0 (0)	1 (6.7)
Carbapenem	0 (0)	0 (0)	1 (12.5)	0 (0)	1 (6.7)
Ceftazidime-Avibactam	0 (0.0)	0 (0)	1 (12.5)	0 (0)	1 (6.7)
Polymyxin/Colistin	1 (25)	0 (0)	0 (0)	0 (0)	1 (6.7)
Dual Therapy	2 (50)	1 (100)	3 (37.5)	1 (50)	7 (46.7)
Carbapenem + Aminoglycoside	1 (25)	0 (0)	0 (0)	0 (0)	1 (6.7)
Carbapenem + Polymyxin/Colistin	0 (0)	0 (0)	1 (12.5)	0 (0)	1 (6.7)
Carbapenem + Tigecycline	0 (0)	0 (0)	1 (12.5)	1 (50)	2 (13.3)
Polymyxin/Colistin + Aminoglycoside	1 (25)	1 (100)	1 (12.5)	0 (0)	3 (20)
Triple Therapy	0 (0)	0 (0)	1 (12.5)	0 (0)	1 (6.7)
Carbapenem + Polymyxin/Colistin + Tigecycline	0 (0)	0 (0)	1 (12.5)	0 (0)	1 (6.7)
4 Drugs or More	0 (0)	0 (0)	2 (25)	0 (0)	2 (13.3)
Carbapenem + Polymyxin/Colistin + Aminoglycoside + Tigecycline	0 (0)	0 (0)	2 (25)	0 (0)	2 (13.3)

*One patient received ceftazidime-avibactam (which was only permitted per protocol as monotherapy) in combination with other antimicrobial agents and is therefore not represented.

- Efficacy among patients with confirmed CRE infections by infection type is summarized in **Table 3** and **Figures 2 and 3**.
- Exploratory risk-benefit analyses were conducted to determine the risk-benefit profile of M-V vs. BAT. Results showed a significant decrease in poor outcomes in patients treated with M-V compared to BAT (mCRE-MITT), as shown in **Table 4**.
 - For the composite endpoint of mortality-nephrotoxicity (defined as either a ≥1.0 mg/dL increase in creatinine or renal AEs of failure, acute failure or impairment), M-V was associated with a reduction in mortality-nephrotoxicity compared to BAT (25.0% vs. 40.0%; 95%CI -44.5% to 14.5%, *P*=.32 and 21.4% vs. 60.0%; 95% CI -67.7% to -9.5%, *P*<.01, respectively).
 - For the composite endpoint of failure-nephrotoxicity (same definitions), M-V was associated with a superior risk-benefit profile compared to BAT (32.1% vs. 80.0%; 95%CI -74.5% to -21.2%, *P*<.001 both definitions).
- Adverse events (AEs) and safety endpoints (safety population/MITT) are shown in **Poster#1862, Kaye K et al, IDWeek 2017**.

Table 3. Efficacy Endpoints by Infection Type Among Patients with Confirmed CRE Infections (mCRE-MITT)

	M-V n/N' (%)	BAT n/N' (%)	Absolute Difference (M-V – BAT)	Relative Difference [(M-V – BAT)/BAT]
Patients with HABP/VABP and bacteremia, combined				
Day-28 all-cause mortality	4/16 (25.0)	4/9 (44.4)	−19.4%	−43.7%
Patients with cUTI/AP				
Overall success rate[a] at EOT	8/11(72.7)	2/4 (50.0)	22.7%	45.4%
Overall success rate[a] at TOC (EOT +7d)[b]	3/7 (42.9)	2/4 (50.0)	−7.1%	−14.0%
Patients with cIAI				
Clinical cure at TOC	1/1 (100)	0/2 (0)	100%	–

[a] Overall success rate was a composite of clinical cure and microbial eradication.
[b] 4 Patients in the M-V group were indeterminate/not assessed at TOC.

Table 4. Exploratory Risk-Benefit Analysis: Patients with Poor Outcomes of Confirmed CRE Infections (mCRE-MITT)

Outcome	M-V (N=28) n (%)	BAT (N=15) n (%)	Absolute Difference (M-V – BAT) (95% CI)	*P* value	Relative Difference [(M-V – BAT)/BAT]
Day-28 All-Cause Mortality or Nephrotoxicity[a]	7 (25.0)	6 (40.0)	-15.0 (-44.5 to 14.5)	.32	-37.5
Clinical Failure or Nephrotoxicity[b]	9 (32.1)	12 (80.0)	-47.9 (-74.5 to -21.2)	<.001	-59.9
Day-28 All-Cause Mortality or Renal AEs[c]	6 (21.4)	9 (60.0)	-38.6 (-67.7 to -9.5)	<.01	-64.3
Clinical Failure or Renal AEs[d]	9 (32.1)	12 (80.0)	-47.9 (-74.5 to -21.2)	<.001	-59.9

[a]Composite outcome of either Day-28 all-cause mortality or a post-baseline increase in serum creatinine ≥1.0 mg/dL.
[b]Composite outcome of either clinical failure at test of cure or a post-baseline increase in serum creatinine ≥1.0 mg/dL.
[c]Composite outcome of either Day-28 all-cause mortality or adverse event of renal failure, renal failure acute or renal impairment.
[d]Composite outcome of either clinical failure at test of cure or adverse event of renal failure, renal failure acute or renal impairment.

Figure 2. Efficacy Endpoints in Patients with HABP/VABP or Bacteremia, by Timepoint (mCRE-MITT)

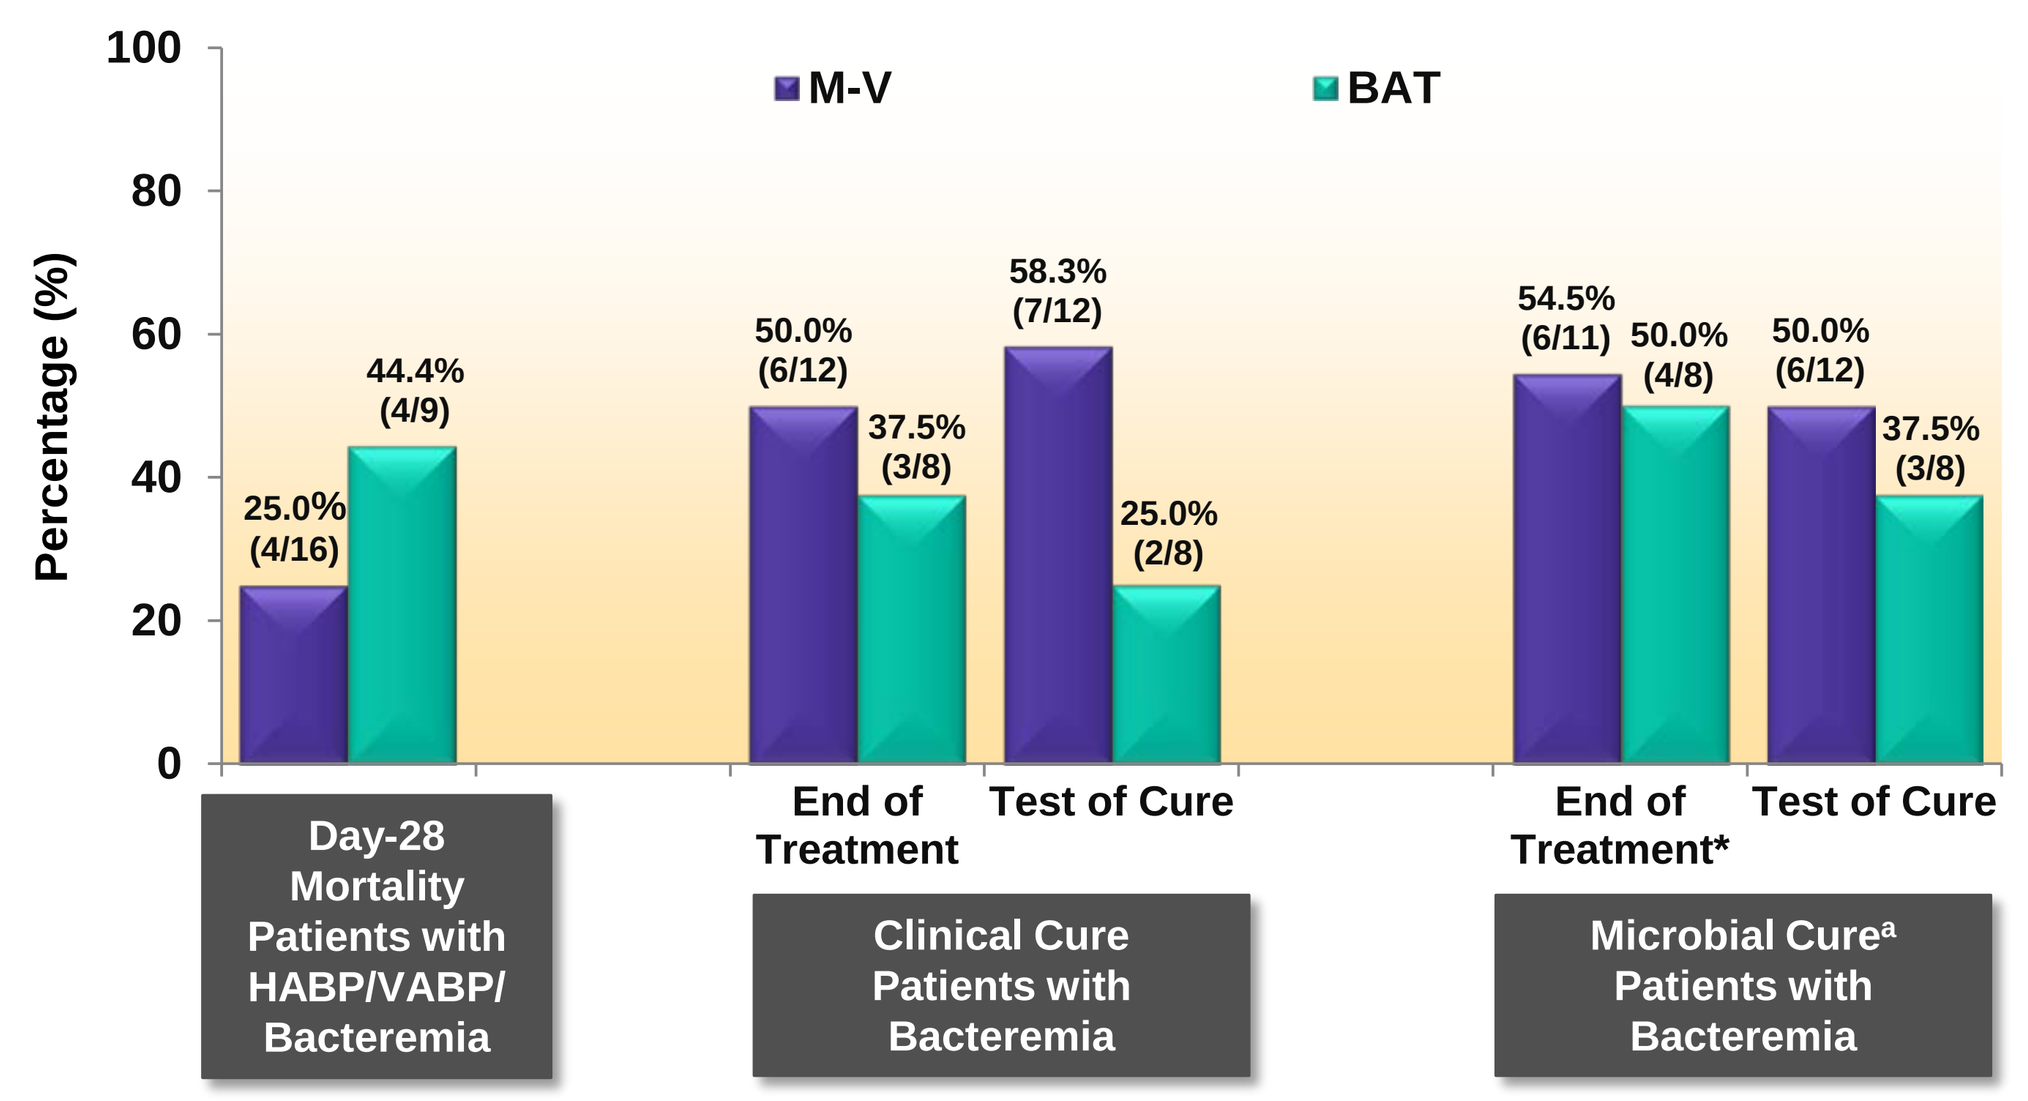
M-V
BAT
Percentage (%)
100
80
60
40
20
0
25.0% (4/16)
44.4% (4/9)
50.0% (6/12)
37.5% (3/8)
58.3% (7/12)
25.0% (2/8)
54.5% (6/11)
50.0% (4/8)
50.0% (6/12)
37.5% (3/8)
End of Treatment
Test of Cure
End of Treatment*
Test of Cure
Day-28 Mortality Patients with HABP/VABP/ Bacteremia
Clinical Cure Patients with Bacteremia
Microbial Cure[a] Patients with Bacteremia

[a] Composite of either microbial eradication or presumed eradication at respective visit.
* One subject in the M-V arm was indeterminate/not assessed at TOC.

Figure 3. Efficacy Endpoints in Patients with cUTI/AP, by Timepoint (mCRE-MITT)

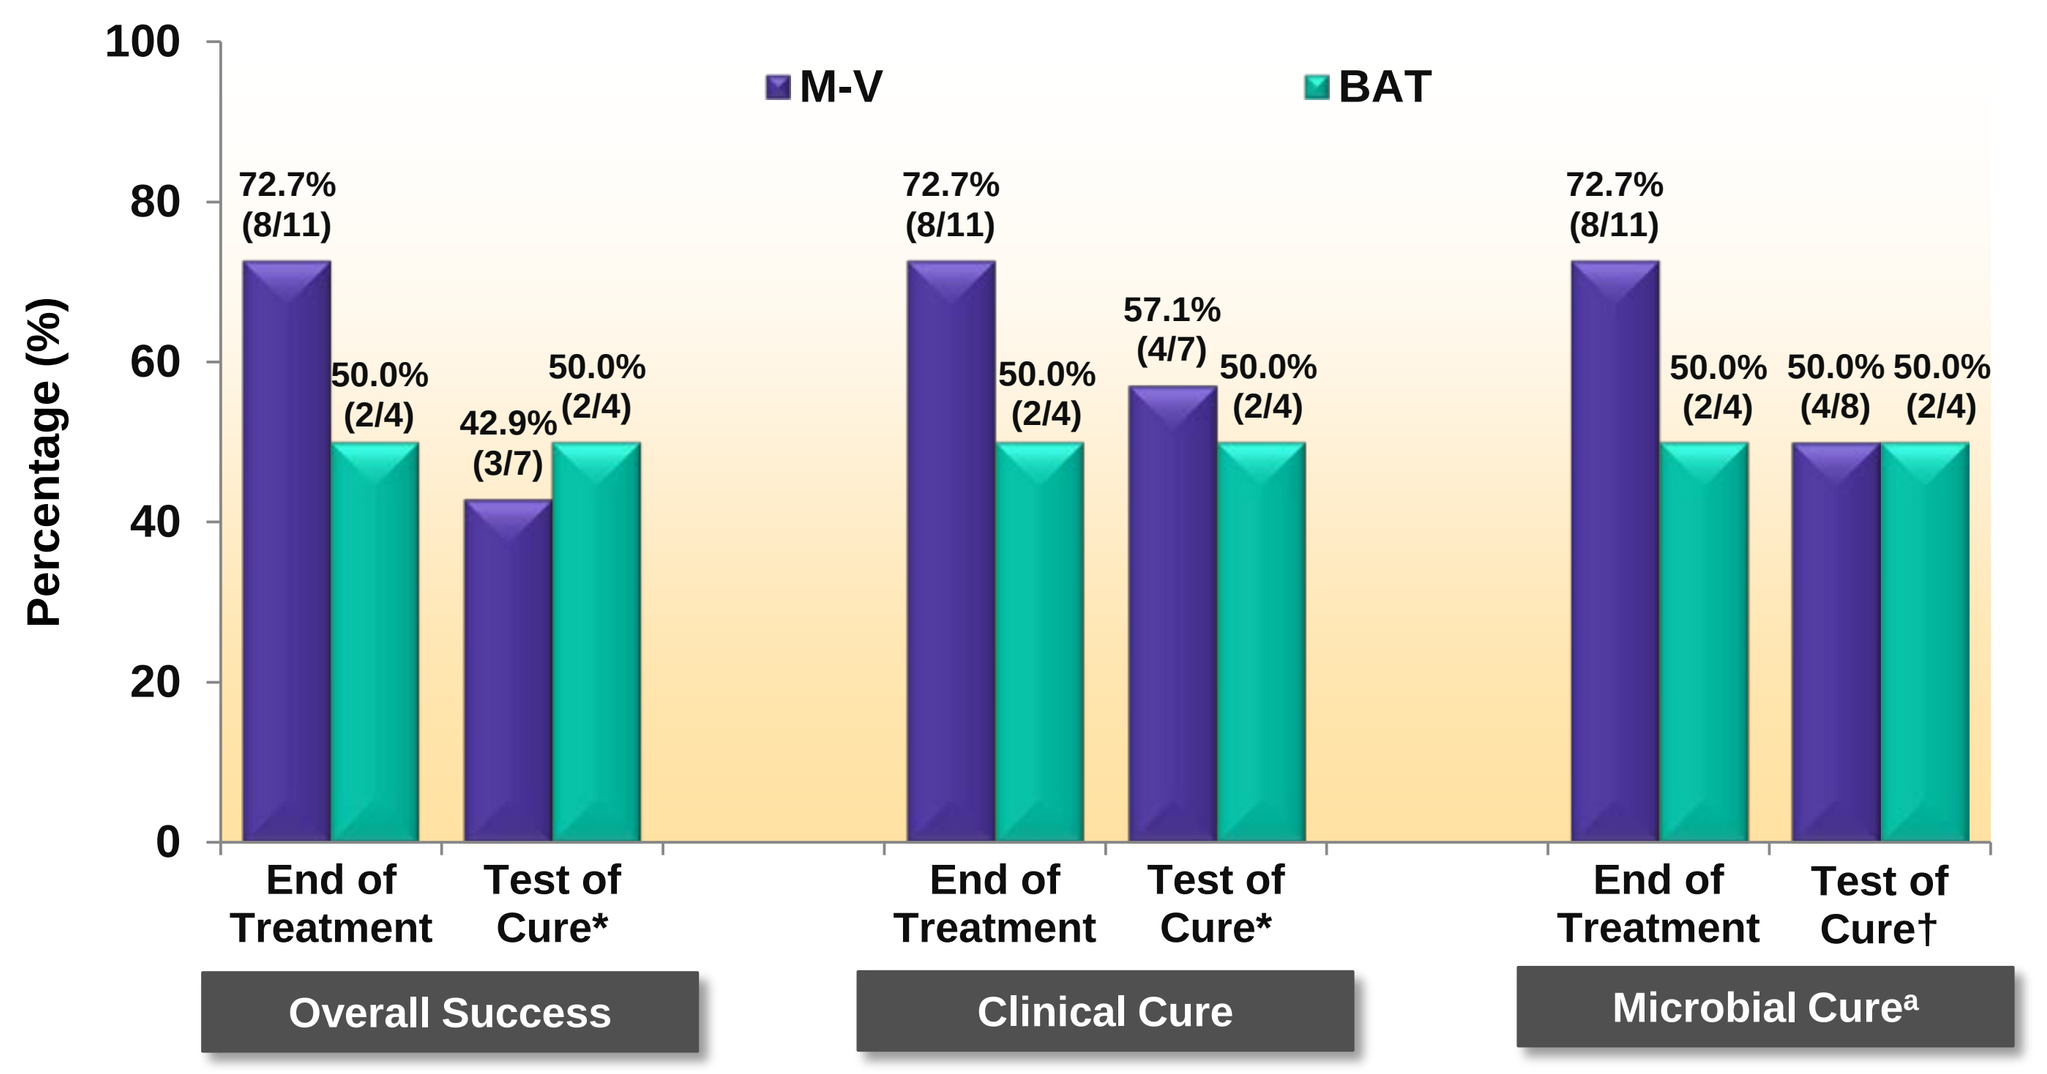
M-V
BAT
Percentage (%)
100
80
60
40
20
0
72.7% (8/11)
50.0% (2/4)
42.9% (3/7)
50.0% (2/4)
72.7% (8/11)
50.0% (2/4)
57.1% (4/7)
50.0% (2/4)
72.7% (8/11)
50.0% (2/4)
50.0% (4/8)
50.0% (2/4)
End of Treatment
Test of Cure*
End of Treatment
Test of Cure*
End of Treatment
Test of Cure†
Overall Success
Clinical Cure
Microbial Cure[a]

[a] Composite of either microbial eradication or presumed eradication at respective visit.
* 4 Patients in the M-V arm were indeterminate/not assessed at TOC.
† 3 Patients in the M-V arm were indeterminate /not assessed at TOC.

Conclusions

Monotherapy with M-V in CRE infections, including immunocompromised, renally impaired, and prior antibiotic failure patients, resulted in the following:

- M-V showed improvement over BAT in efficacy endpoints in all infection types studied, including HABP/VABP, BSI, cUTI, and cIAI.
- M-V demonstrated a lower rate of renal-related adverse events and increases in serum creatinine compared to BAT (**Poster #1879**).
- M-V was associated with reduction in poor outcomes compared to BAT including failure-nephrotoxicity and mortality-nephrotoxicity on exploratory risk-benefit analyses across all infection types combined.
- M-V is a potent new option for treatment of KPC-driven carbapenem resistance.
- Based on the totality of these results, the independent Data Safety Monitoring Board recommended discontinuing randomization to BAT.

Disclosures

This project has been funded in whole or in part with federal funds from the Department of Health and Human Services; Office of the Assistant Secretary for Preparedness and Response; Biomedical Advanced Research and Development Authority (BARDA), under Contract No. HHSO100201400002C with Rempex Pharmaceuticals, a wholly-owned subsidiary of The Medicines Company and Agreement No. HHSO100201600026C with The Medicines Company.

References

1. Centers for Disease Control and Prevention. Antibiotic resistance threats in the United States, 2013. Atlanta, GA (US): US Department of Health and Human Services; 2013. 114 p.
2. World Health Organization. Antimicrobial resistance: global report on surveillance 2014 [Internet]. Geneva, CH: World Health Organization; 2014 [cited 2017 Jul 31]. Available from: http://apps.who.int/iris/bitstream/10665/112642/1/9789241564748_eng.pdf.
3. Castanheira M, Sader HS, Deshpande LM, Fritsche TR, Jones RN. Antimicrobial activities of tigecycline and other broad-spectrum antimicrobials tested against serine carbapenemase- and metallo-β-lactamase-producing Enterobacteriaceae: report from the SENTRY Antimicrobial Surveillance Program. Antimicrob Agents Chemother. 2008 Feb;52(2):570-3.

Presented at IDWeek 2017, October 4-8, 2017, San Diego, CA