1835

Meropenem-Vaborbactam (VABOMERE) Pharmacokinetics in Subjects with Chronic Renal Impairment, Including Hemodialysis

Christopher M. Rubino[1], David C. Griffith[2], Sujata M. Bhavnani[1], Jeffery S. Loutit[2], Brooke Lohse[2], Michael N. Dudley[2], Paul G. Ambrose[1]

[1]Institute for Clinical Pharmacodynamics, Schenectady, NY, United States; [2]The Medicines Company, San Diego, CA, United States

The Medicines Company
3013 Science Park Rd.
San Diego, CA 92121 USA
8 Sylvan Way
Parsippany, NJ 07054 USA

Abstract

Introduction

Vaborbactam is a member of a new class of beta-lactamase inhibitors with inhibitory activity against important beta-lactamases, particularly serine carbapenemases, and is being developed in combination with meropenem. The combination is highly active against gram-negative pathogens, especially KPC-producing, carbapenem-resistant Enterobacteriaceae. This report describes the pharmacokinetic data of the combination in subjects with chronic renal impairment, including hemodialysis.

Methods

A total of 41 subjects were enrolled in a Phase 1 study and assigned to one of five groups based on renal function. Subjects were classified as having mild, moderate, or severe renal impairment if they had an estimated glomerular filtration rate (eGFR) of 60 to 89, 30 to <60, or <30 mL/min/1.73 m^2, respectively. Normal renal function was defined as an eGFR ≥90 mL/min/1.73 m^2. Subjects with end-stage renal disease (ESRD) who were receiving hemodialysis therapy 3 times a week for at least 3 months prior to Day 1 of the study were the final group. All subjects received a single IV dose of meropenem 1 g - vaborbactam 1 g in combination as a 3-hour infusion. Subjects were dosed on Day 1 with the exception of the ESRD group that received two doses (On- and Off-dialysis) separated by a washout. Plasma samples were assayed for meropenem and vaborbactam using validated HPLC/MS methods; total plasma clearance (CLt) was estimated by standard
non-compartmental methods.

Results

Meropenem and vaborbactam CLt were similar and both drugs are removed by hemodialysis (**Table Below**). Meropenem and vaborbactam CLt decreased with decreasing renal function and the slopes of the relationships between eGFR and CLt were similar (**Figure 2B**), indicating a similar proportional reduction in CLt with decreasing renal function.

Mean (CV) clearance by renal function category

eGFR, mL/min/1.73 m^2	CLt (L/h)	
	Meropenem	Vaborbactam
eGFR ≥90	12.0 (31.9%)	10.6 (34.3%)
eGFR 60-89	9.33 (30.5%)	8.94 (26.3%)
eGFR 30-59	5.80 (32.9%)	4.56 (44.2%)
eGFR <30	2.59 (24.3%)	1.35 (36.1%)
ESRD Off-Dialysis	1.66 (31.6%)	0.282 (127%)
ESRD On-Dialysis	3.65 (21.1%)	1.04 (62.7%)

Conclusion

These data suggest that dose reduction in subjects with renal impairment should be similar for both meropenem and vaborbactam. Along with the population pharmacokinetic data from the TANGO studies, these data provided a basis for the USPI recommended dose adjustment for patients with renal impairment (**Table 3**).[1]

Background

- Vaborbactam is a member of a new class of beta-lactamase inhibitors with inhibitory activity against important beta-lactamases, particularly serine carbapenemases. Vaborbactam is being developed in combination with meropenem for the treatment of patients with severe gram-negative infections.
- The purpose of this Phase 1 study was to assess the safety, tolerability, and pharmacokinetics of meropenem-vaborbactam (M-V; VABOMERE) administered to adult subjects with renal impairment compared with subjects with normal renal function.

Methods

- Pharmacokinetics of meropenem and vaborbactam were assessed in a Phase 1, open-label, single-dose study of adult subjects with varying degrees of renal impairment.
- A single dose of meropenem 1 g and vaborbactam 1 g was chosen for this study.
- 41 subjects were enrolled and formed 5 study groups based on renal function:
 - Group 1: mild renal impairment (estimated glomerular filtration rate [eGFR] of 60 to 89 mL/min/1.73 m^2)
 - Group 2: moderate renal impairment (eGFR of 30 to <60 mL/min/1.73 m^2)
 - Group 3: severe renal impairment (eGFR <30 mL/min/1.73 m^2)
 - Group 4: normal renal function (eGFR >90 mL/min/1.73 m^2)
 - Group 5: subjects with end-stage renal disease (ESRD) receiving hemodialysis (HD) therapy 3 times/week at least 3 months prior to Day 1 of study
 - Group 5 received 2 doses (on- and off-dialysis)
- For all renal function groups, blood samples were collected for pharmacokinetic assessments.
 - For Groups 1-4, urine samples were also collected
 - For Group 5, dialysate was collected prior to initiation of HD and hourly during the HD session

Results

Demographics

- Summary of baseline subject demographics are found in **Table 1.**
 - Majority of subjects were white males. Ages ranged from 43 to 73 years

Pharmacokinetics

- A summary of pharmacokinetic parameters by renal function group is found in **Table 2**.
- Concentrations of meropenem and vaborbactam increased over time with decreasing renal function. (**Figure 1A** and **1B**, respectively).
 - For meropenem, the mean area under the concentration-time curve from time zero to infinity (AUC0-inf) increased from 87.1 µg • h/mL in subjects with normal renal function to:
 - 112 µg • h/mL in subjects with mild impairment
 - 181 µg • h/mL in subjects with moderate impairment
 - 397 µg • h/mL in subjects with severe impairment
 - 629 µg • h/mL in subjects with ESRD in between dialysis sessions (Off-dialysis)
- Similar trend observed for vaborbactam, but increase in $AUC_{0\text{-inf}}$ was more pronounced as renal function decreased.
 - For vaborbactam, the mean AUC0-inf increased from 99.4 µg • h/mL in subjects with normal renal function to:
 - 116 µg • h/mL in subjects with mild impairment
 - 238 µg • h/mL in subjects with moderate impairment
 - 781 µg • h/mL in subjects with severe renal impairment
 - 5220 µg • h/mL in subjects with ESRD between dialysis sessions (Off-dialysis)
- Group 5 had a noticeable increase in drug clearance with dialysis.
 - For meropenem, mean $AUC_{0\text{-inf}}$ was 2.24 times higher for Off-dialysis vs. On-dialysis
 - For vaborbactam, mean AUC0-inf was nearly 5 times higher Off-dialysis vs. On-dialysis
- Statistically significant relationships were found between eGFR and renal clearance (CLr) and total plasma clearance (CLt) for both compounds:
 - The slopes for the relationships between CLt and eGFR were nearly identical (**Figure 2**)
 - The slope of the relationship between vaborbactam CLr and eGFR was steeper than that for meropenem CLr and eGFR
- Impact of HD on clearance of M-V was evaluated in Group 5.
 - HD performed immediately following drug administration significantly increased drug clearance of both meropenem and vaborbactam
 - Mean increase in CLt was 2.21-fold for meropenem and 5.11-fold for vaborbactam (**Figure 3**)
- The median percentage of dose recovered in dialysate during the HD session was 38.3% and 52.9% for meropenem and vaborbactam, respectively.
- Dosage adjustment is recommended in patients with renal impairment who have an eGFR less than 50 mL/min/1.73m^2 (**Table 3**).[1]

Safety

- 14 subjects (34.1%) reported a total of 20 treatment emergent adverse events (TEAEs) during the study.
- No increase in the incidence of adverse events (AEs) was observed with decreasing renal function; however, an increased number of subjects reported AEs in the ESRD group in the period when M-V was given after dialysis as opposed to before (63% vs. 22%).
- Most frequent TEAEs were diarrhea, headache, abdominal pain, and dermatitis.
- Majority of AEs were mild.
- Two severe AEs were reported in subjects with ESRD, one of metastatic prostate cancer and one of hemorrhagic diarrhea.

Table 1. Baseline demographics

Parameter	Normal (n=8)	Mild (n=8)	Moderate (n=8)	Severe (n=8)	ESRD (n=9)	Total (n=41)
		Renal Impairment				
Sex (n [%])						
Female	4 (50)	4 (50)	4 (50)	3 (37.5)		15 (36.6)
Male	4 (50)	4 (50)	4 (50)	5 (62.5)	9 (100)	26 (63.4)
Race (n [%])						
White	7 (87.5)	5 (62.5)	5 (62.5)	2 (25.0)	3 (33.3)	22 (53.7)
Black of African American		3 (37.5)	2 (25.0)	5 (62.5)	5 (55.6)	15 (36.6)
Other	1 (12.5)		1 (12.5)	1 (12.5)	1 (11.1)	4 (9.8)
Ethnicity (n [%])						
Hispanic or Latino	1 (12.5)	1 (12.5)	1 (12.5)		2 (22.2)	5 (12.2)
Not Hispanic or Latino	7 (87.5)	7 (87.5)	7 (87.5)	8 (100)	7 (77.8)	36 (87.8)
Age (yrs)						
Mean (SD)	56.8 (3.4)	53.4 (8.9)	62.1 (10.2)	57.6 (6.4)	53.8 (10.5)	56.7 (8.6)
Range	52–62	44–67	45–73	49–69	43–73	43–73
Height (cm)						
Mean (SD)	169.6 (9.1)	172.9 (9.4)	170.9 (8.1)	174.4 (9.6)	177.8 (4.6)	173.2 (8.4)
Range	160–187	156.3–189.6	161.3–183.4	159.7–186.5	172–186	156.3–189.6
Weight (kg)						
Mean (SD)	91.6 (11.0)	99.4 (22.1)	89.2 (21.4)	94.2 (19.4)	106.0 (12.9)	96.4 (18.0)
Range	77.5–107.8	67.5–142.6	61.1–122.7	74.1–125.7	85.6–121.6	61.1–142.6
BMI (kg/m^2)						
Mean (SD)	31.8 (1.8)	33.1 (5.5)	30.4 (5.9)	31.1 (6.7)	33.6 (4.2)	32.0 (5.0)
Range	30.3–34.7	21.5–39.7	22.7–40.7	26.2–43.8	28.0–40.1	21.5–43.8
eGFR[a] (mL/min/m^2)[a]						
Mean (SD)	99.6 (23.4)	75.3 (6.2)	44.1 (8.2)	15.1 (4.8)		47.3 (37.3)
Range	66–141	67–83	32–56	10–25		5–141

[a]eGFR was calculated using the Modification of Diet in Renal Disease (MDRD) equation. Assignment to renal function groups was based on eGFR values at screening; baseline eGFR values may differ from screening values.

Figure 1. Plots of median meropenem (A) and vaborbactam (B) concentration-time profiles, stratified by renal function group and timing relative to dialysis (for ESRD subjects)

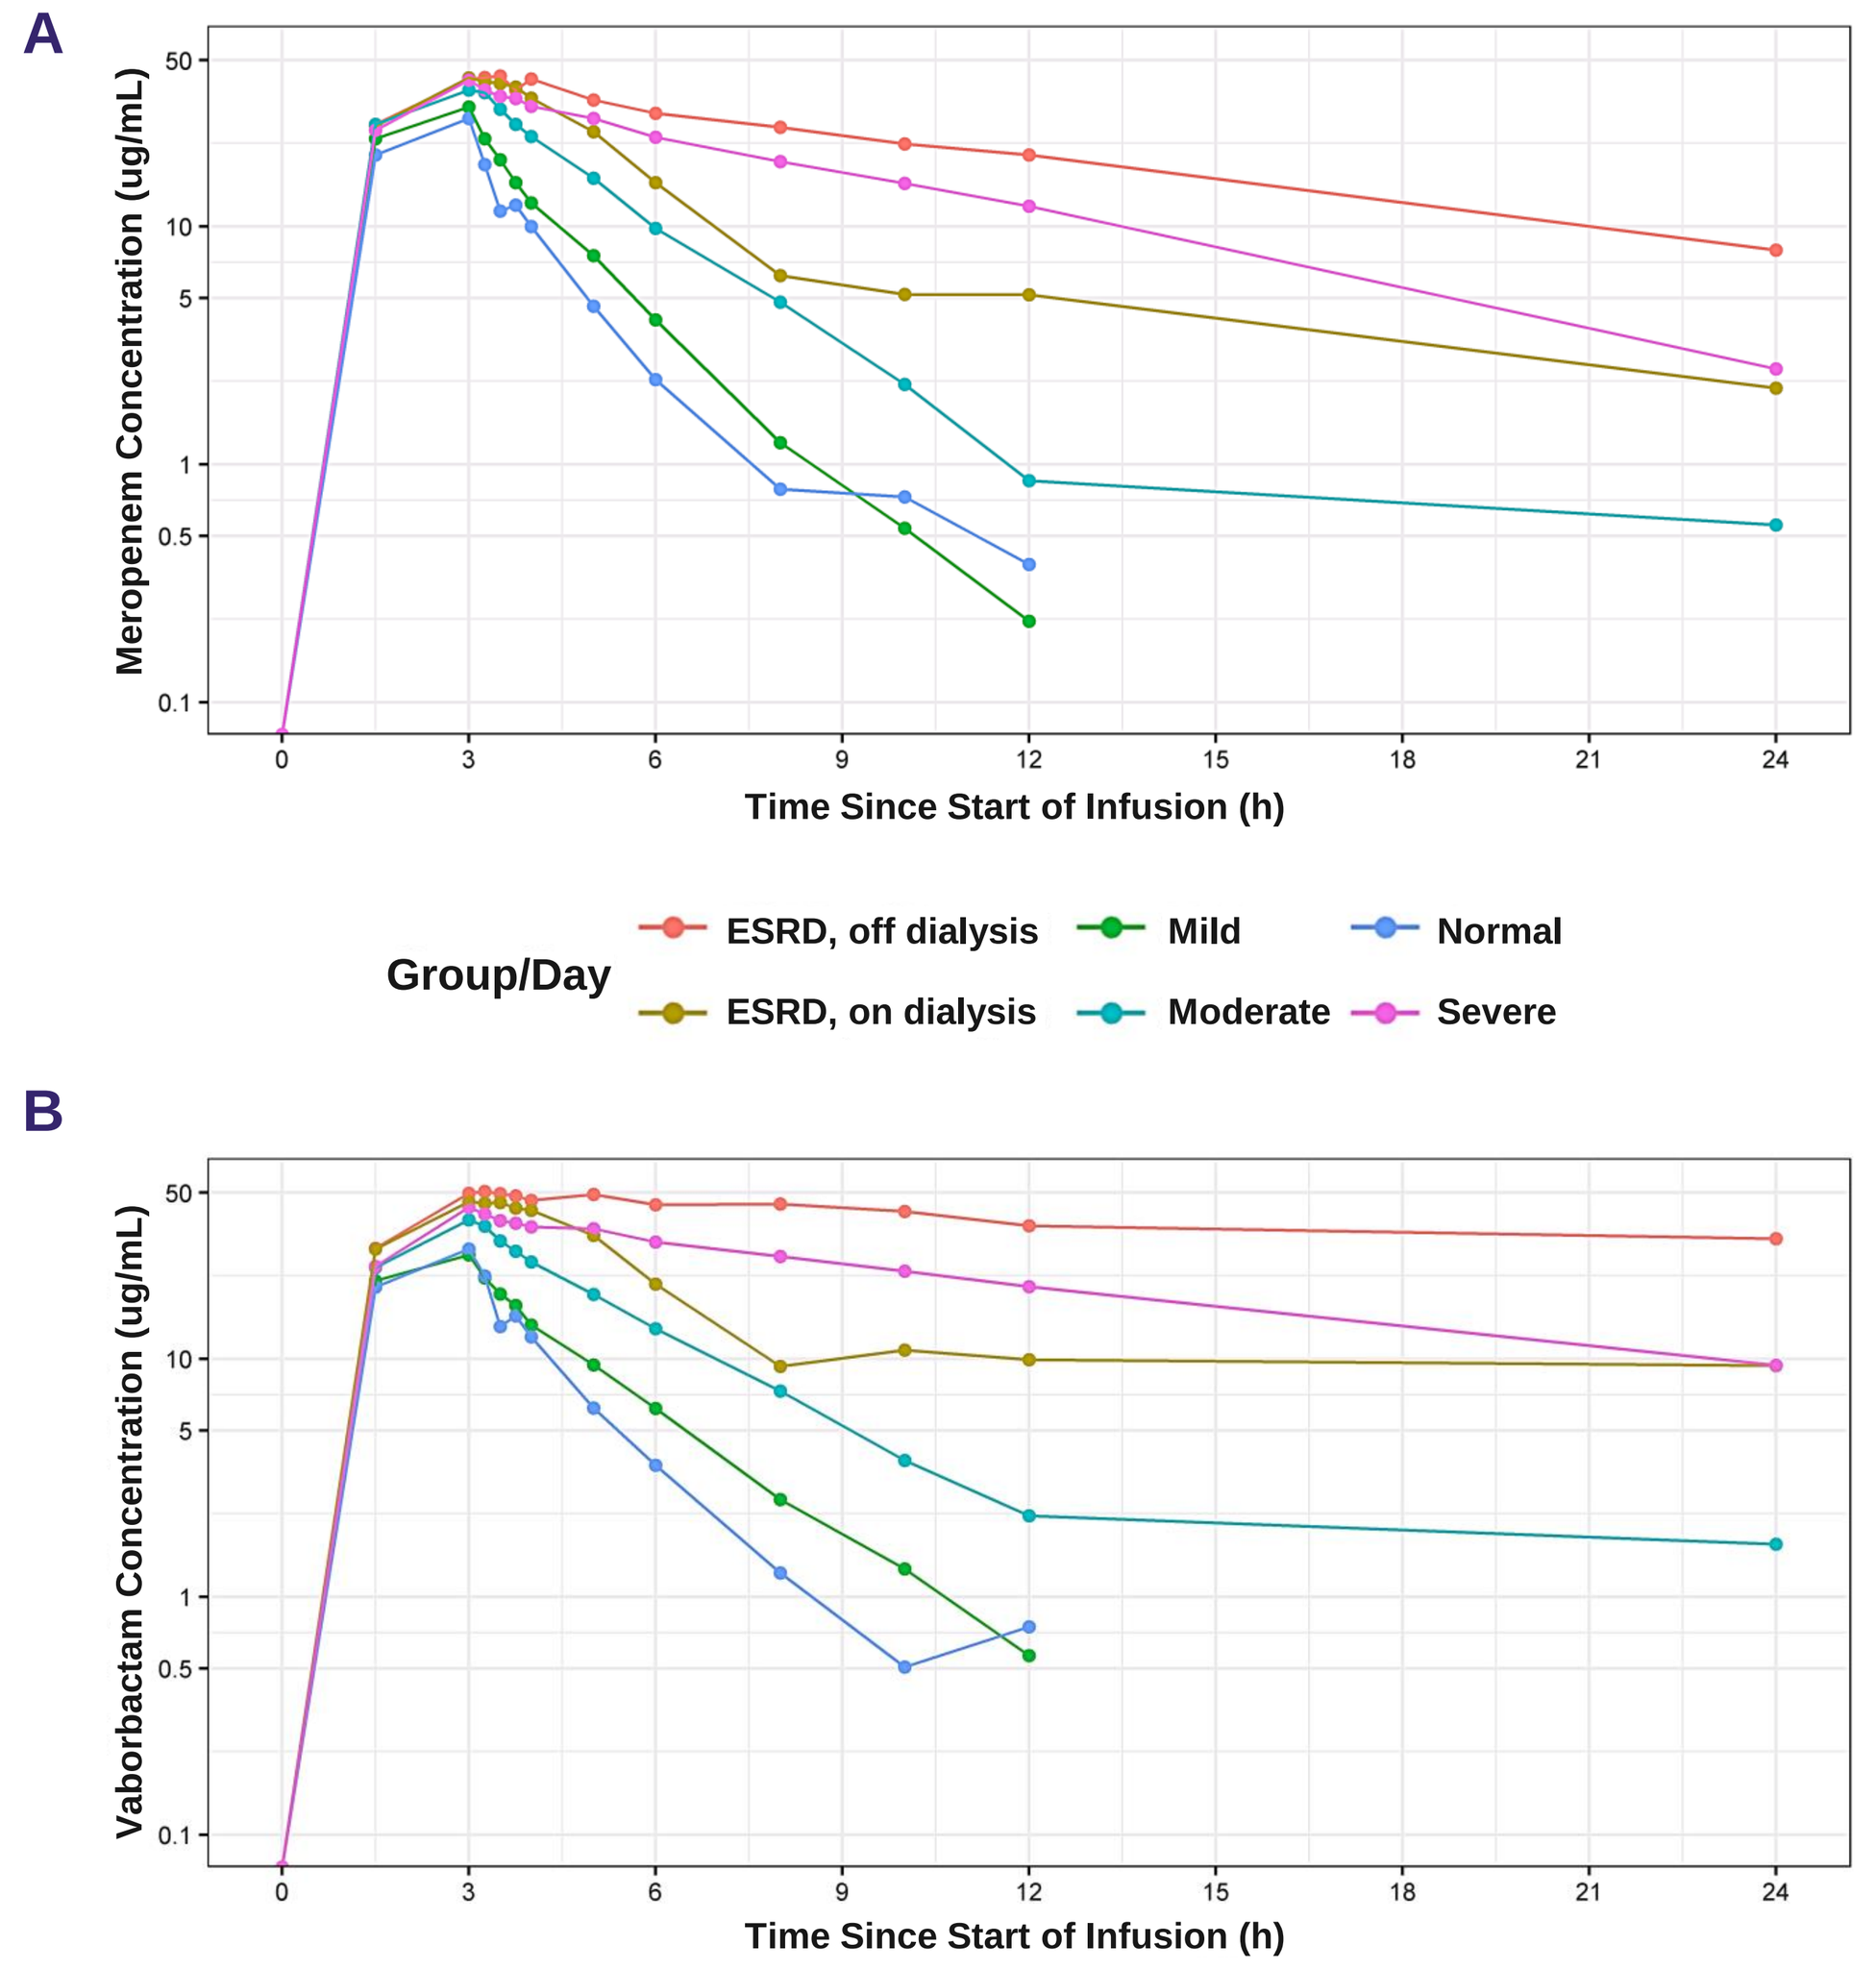

A
Meropenem Concentration (ug/mL)
Time Since Start of Infusion (h)
Group/Day
ESRD, off dialysis
ESRD, on dialysis
Mild
Moderate
Normal
Severe
B
Vaborbactam Concentration (ug/mL)
Time Since Start of Infusion (h)

Table 2. Pharmacokinetic parameters of meropenem and vaborbactam stratified by renal function groupa

Renal Study Group	C_{max} (µg/mL)	$AUC_{0\text{-inf}}$ (µg • h/mL)	$AUC_{0\text{-t}}$ (µg • h/mL)	$t_{1/2}$ (h)	CL_{NR} (L/h)	CL_R (L/h)	CL_t (L/h)	Ae (mg)	f_e (%)	V_{ss} (mL)
Pharmacokinetic values for meropenem										
Normal (n=8)	27.5 ± 7.03	87.1 ± 26.7	86.4 ± 26.6	1.30 ± 0.254	4.79 ± 2.18	7.69 ± 2.33	12.5 ± 3.83	625 ± 89.7	62.5 ± 8.97	19.2 ± 2.69
Mild (n=8)	32.7 ± 9.1	112 ± 40.6	111 ± 40.1	1.43 ± 0.179	4.11 ± 1.30	5.55 ± 2.06	9.66 ± 2.42	568 ± 106	56.8 ± 10.6	16.5 ± 3.22
Moderate (n=8)	40.5 ± 6.27	181 ± 59.4	177 ± 57.2	2.19 ± 0.867	2.71 ± 0.811	3.34 ± 1.19	6.05 ± 1.83	547 ± 5.98	54.7 ± 5.98	16.6 ± 2.21
Severe (n=8)	45.5 ± 13.0	397 ± 98.0	362 ± 83.3	6.10 ± 2.59	1.69 ± 0.376	0.964 ± 0.375	2.65 ± 0.606	355 ± 86.1	35.5 ± 8.61	21.6 ± 6.69
ESRD Off-dialysis (n=8)	44.7 ± 8.40	280 ± 58.7	274 ± 56.8	9.33 ± 1.94	NA	NA	3.72 ± 0.764	378 ± 45.7	37.8 ± 4.57	22.2 ± 6.33
ESRD On-dialysis (n=9)	47.7 ± 11.7	629 ± 206	606 ± 196	9.45 ± 1.75	NA	NA	1.73 ± 0.512	NA	NA	30 ± 7.18
Pharmacokinetic values for vaborbactam										
Normal (n=8)	27.8 ± 6.34	99.4 ± 33.0	98.0 ± 32.2	1.65 ± 0.352	0.554 ± 1.69	10.5 ± 2.77	11.1 ± 3.60	972 ± 122	97.2 ± 12.2	20.5 ± 3.53
Mild (n=8)	30.1 ± 8.31	116 ± 35.9	114 ± 34.5	1.88 ± 0.274	1.67 ± 1.52	7.51 ± 1.99	9.17 ± 1.98	825 ± 144	82.5 ± 14.4	20.8 ± 3.73
Moderate (n=8)	42.4 ± 7.45	238 ± 108	229 ± 99.9	3.45 ± 1.71	0.642 ± 0.402	4.25 ± 1.66	4.89 ± 1.78	860 ± 73.3	86.0 ± 7.33	18.8 ± 3.02
Severe (n=8)	46.4 ±12.3	781 ± 279	532 ± 110	13.5 ± 8.71	0.288 ± 0.271	1.13 ± 0.505	1.42 ± 0.465	714 ± 190	71.4 ± 19.0	23.3 ± 7.24
ESRD Off-dialysis (n=8)	56.5 ± 13.9	5220 ± 4620	1640 ± 567	79.3 ± 76.2	NA	NA	0.405 ± 0.329	NA	NA	22.2 ± 5.47
ESRD On-dialysis (n=9)	50.1 ± 10.2	1100 ± 539	533 ± 124	55.2 ± 33.6	NA	NA	1.22 ± 0.799	528 ± 49.1	52.8 ± 4.91	59.1 ± 16.8

[a]Values shown are means ± SD

Figure 2. Relationship between eGFR and CLt of meropenem and vaborbactam

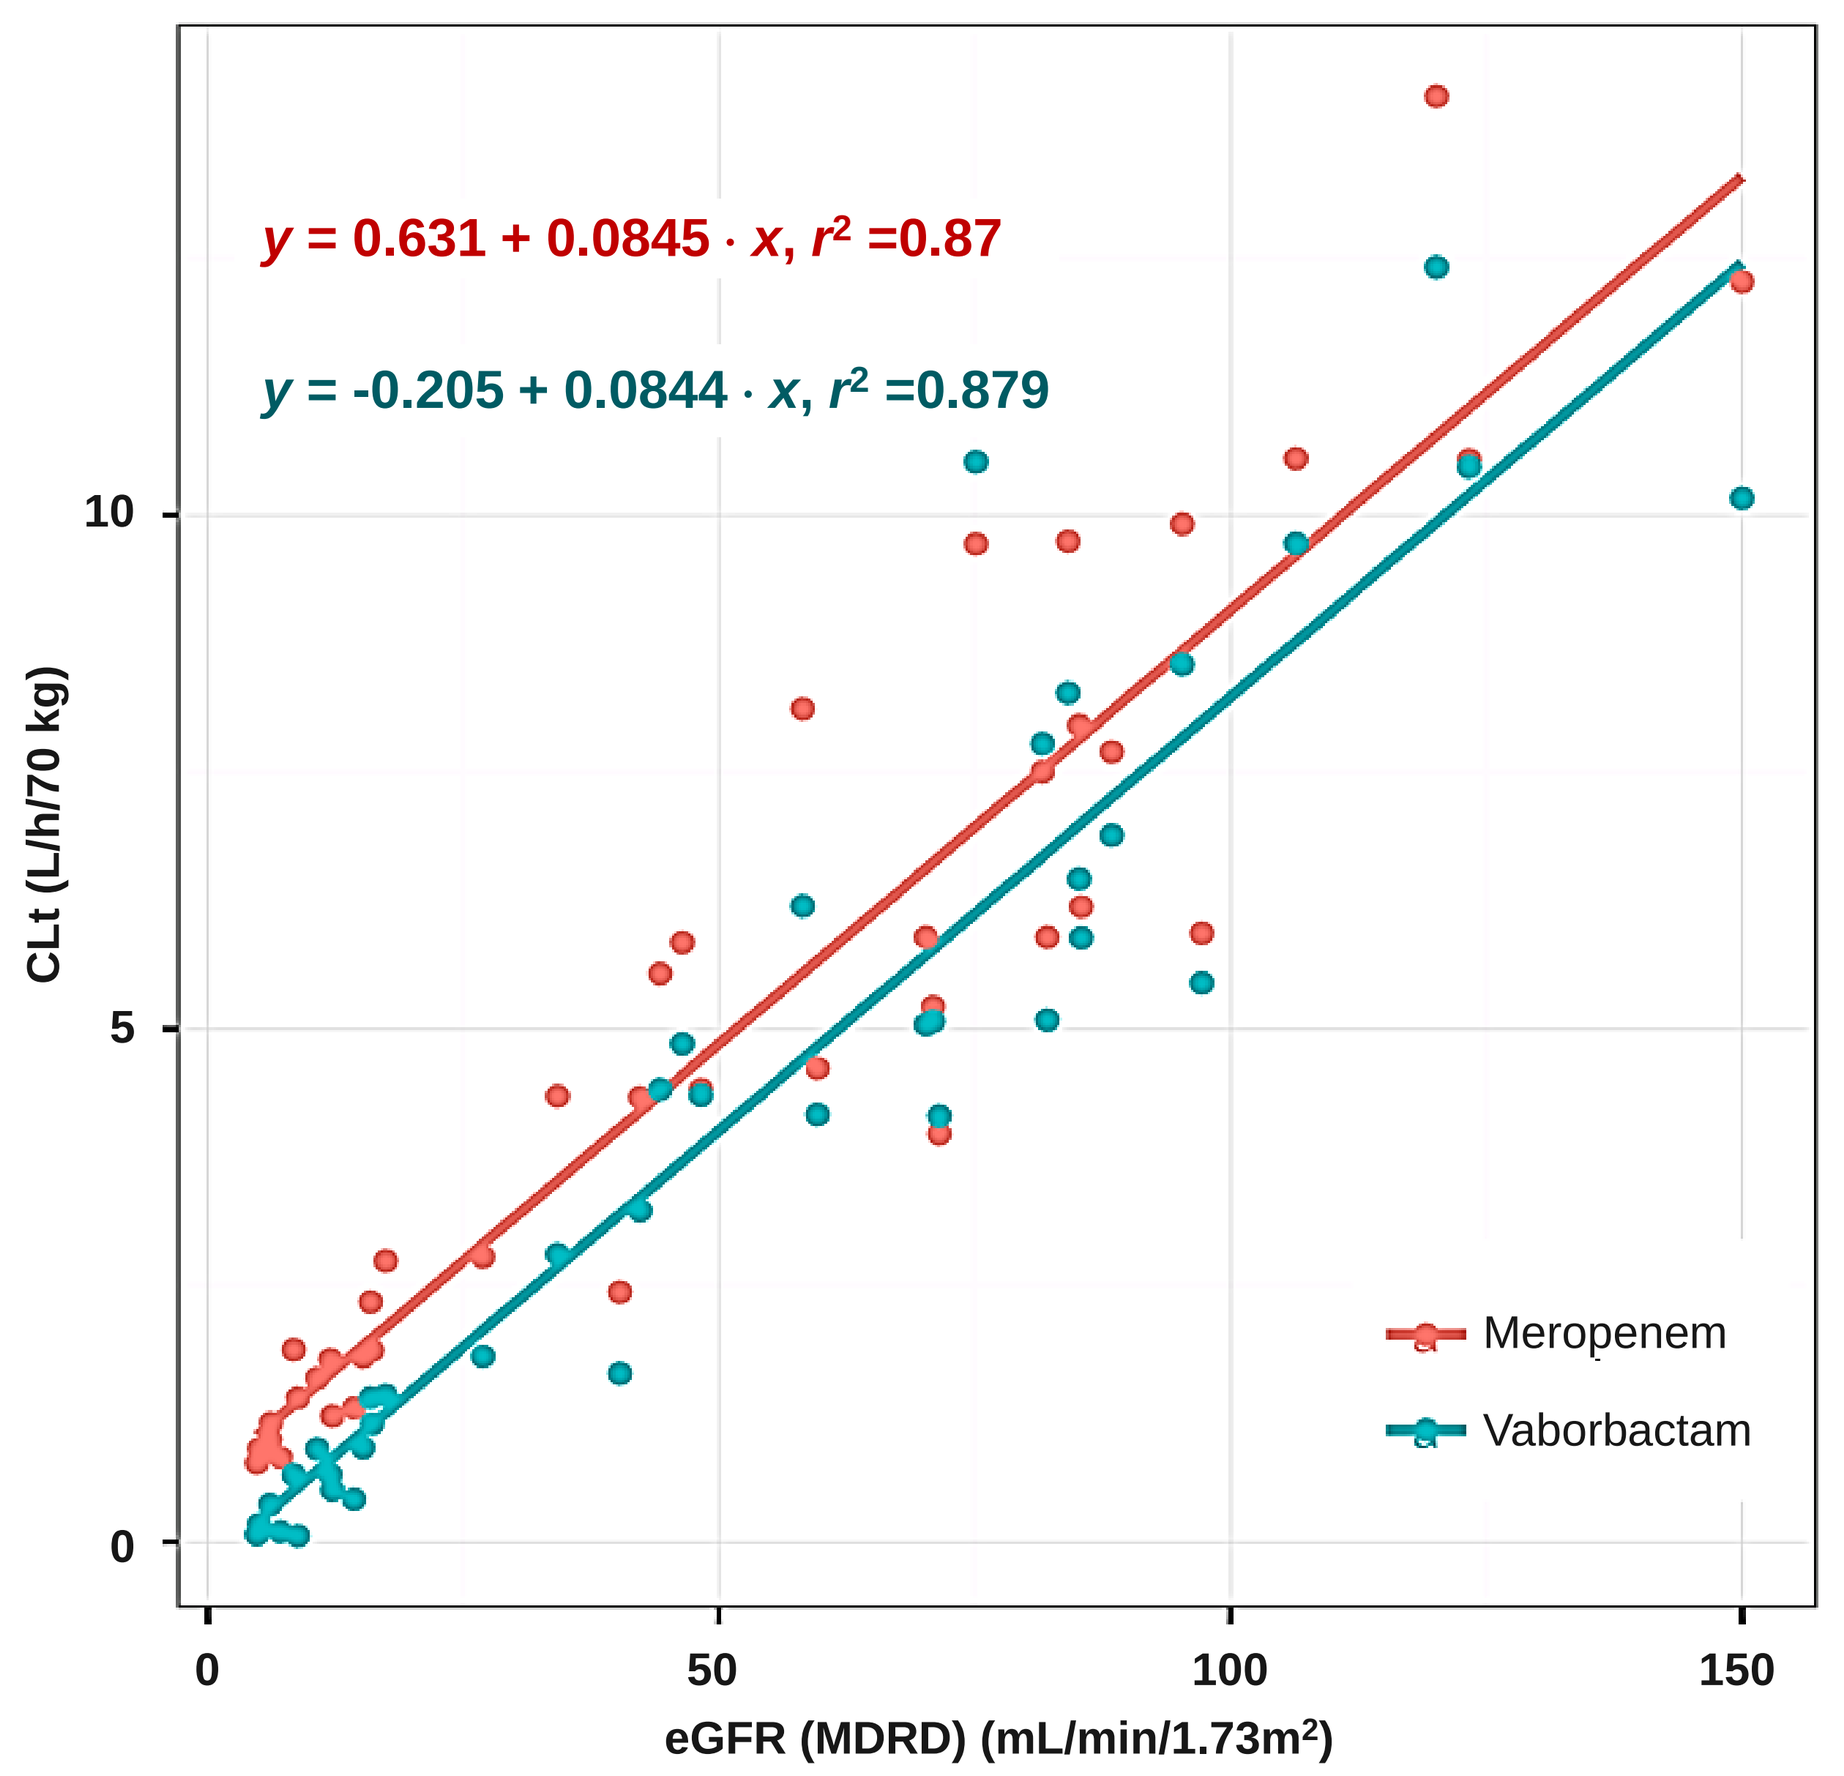

y = 0.631 + 0.0845 · x, r² =0.87
y = -0.205 + 0.0844 · x, r² =0.879
CLt (L/h/70 kg)
eGFR (MDRD) (mL/min/1.73m²)
Meropenem
Vaborbactam

Figure 3. Effect of HD on CLt of meropenem and vaborbactam patients with ESRD

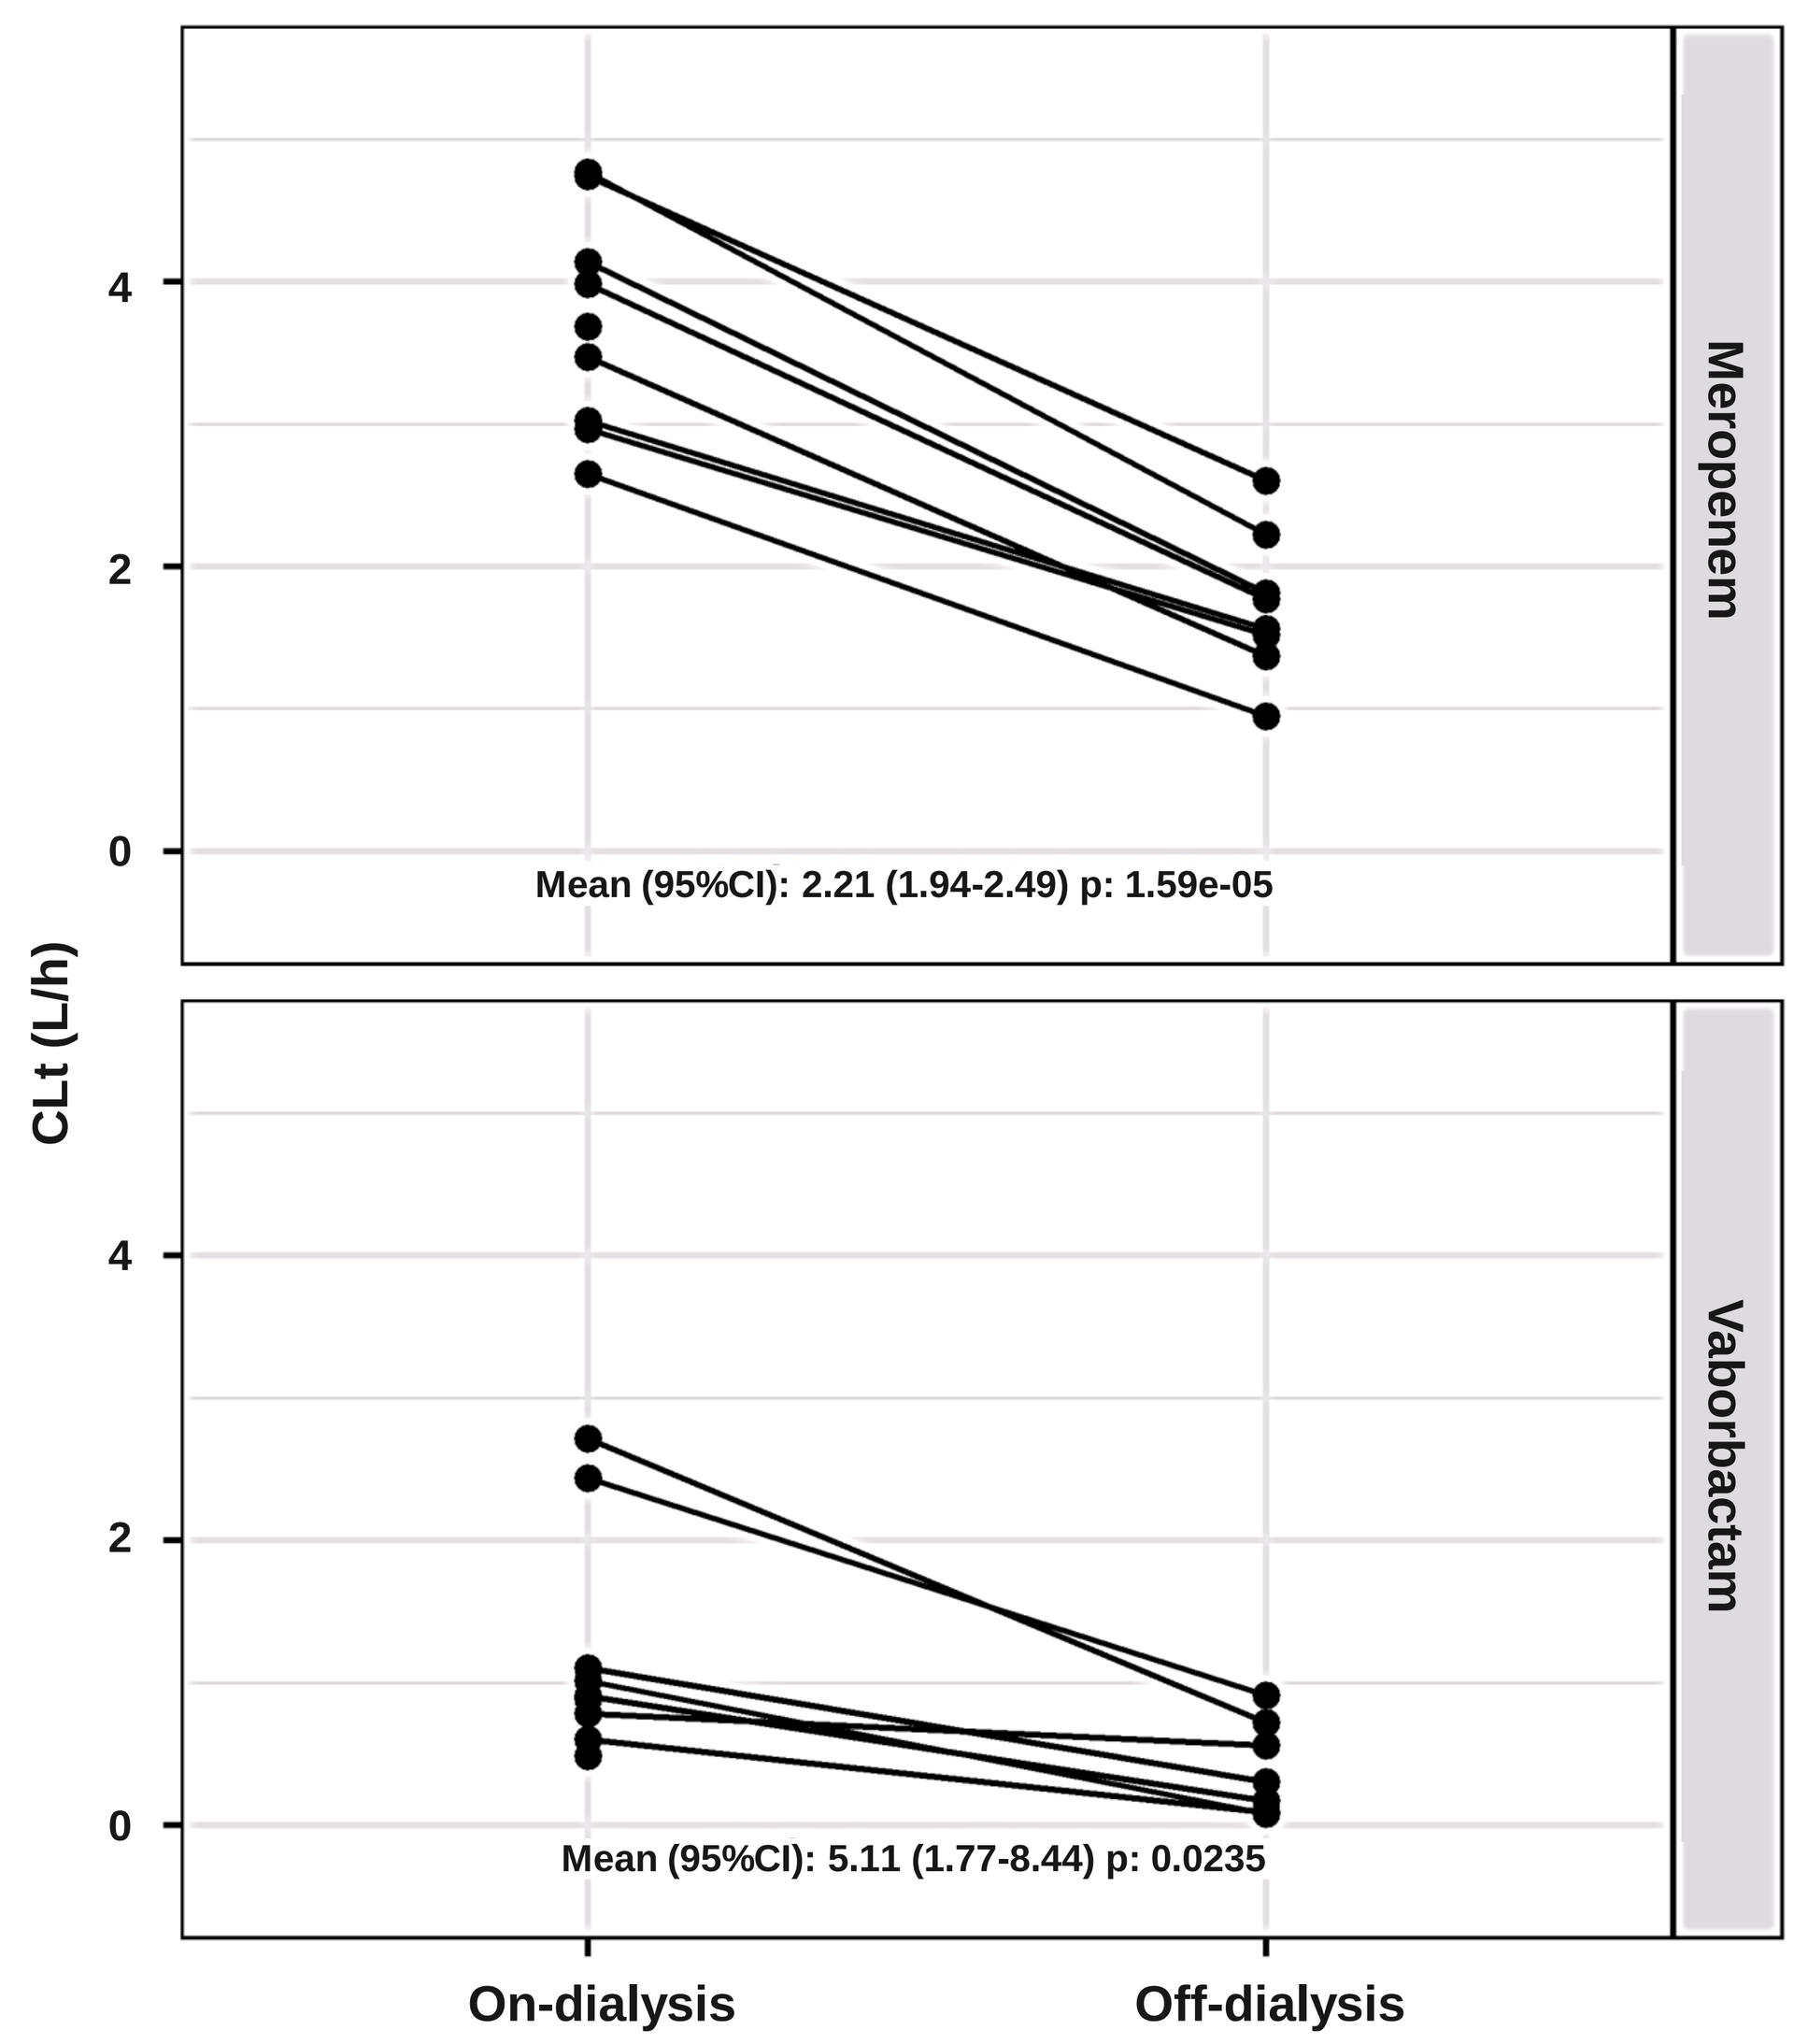

CLt (L/h)
Meropenem
Vaborbactam
Mean (95%CI): 2.21 (1.94-2.49) p: 1.59e-05
Mean (95%CI): 5.11 (1.77-8.44) p: 0.0235
On-dialysis
Off-dialysis

Table 3. Recommended dose adjustment in patients with renal impairment[1]

eGFR[a] (mL/min/ 1.73m^2)	Recommended Dosage Regimen for VABOMERE (meropenem and vaborbactam)[b, c, d]	Dosing Interval
30 to 49	VABOMERE 2 grams (meropenem 1 gram and vaborbactam 1 gram)	Every 8 hours
15 to 29	VABOMERE 2 grams (meropenem 1 gram and vaborbactam 1 gram)	Every 12 hours
Less than 15	VABOMERE 1 gram (meropenem 0.5 grams and vaborbactam 0.5 grams)	Every 12 hours

[a]As calculated using the Modification of Diet in Renal Disease (MDRD) formula.
[b]All doses of VABOMERE are administered intravenously over 3 hours.
[c]Doses adjusted for renal impairment should be administered after a hemodialysis session.
[d]The total duration of treatment is for up to 14 days.

Conclusions

- Meropenem and vaborbactam are both highly renally cleared, and drug concentrations increase with worsening renal function, requiring dose adjustments for patients with renal impairment.
- The PK profiles for meropenem and vaborbactam are parallel and produce predictable and proportional exposures for both components even in renally impaired patients.
- Both meropenem and vaborbactam are removed by hemodialysis. A maintenance dose of M-V should be administered after a hemodialysis session.
- M-V is well tolerated in renally impaired patients with no evidence of increasing incidence or severity of AEs.
- In summary, M-V is a safe drug for treatment of serious gram negative infections in renally impaired patients.

Disclosures

This project has been funded in whole or in part with federal funds from the Department of Health and Human Services; Office of the Assistant Secretary for Preparedness and Response; Biomedical Advanced Research and Development Authority (BARDA), under Contract No.HHSO100201400002C with Rempex Pharmaceuticals, a wholly-owned subsidiary of The Medicines Company. ClinicalTrials.gov Identifier: NCT02020434

References

1. VABOMERE [prescribing information]. Parsippany, NJ: The Medicines Company; 2017.

Presented at IDWeek 2017, October 4-8, 2017, San Diego, CA