Meropenem-Vaborbactam (VABOMERE) vs. Best Available Therapy for Carbapenem-Resistant Enterobacteriaceae Infections in TANGO II: Outcomes in Immunocompromised Patients

1868

<u>David Paterson</u>[1], Eun Jeong Kwak[2], Tanaya Bhowmick[3], Elizabeth Alexander[4], Jeff Loutit[5], Shu Zhang[4], Michael Dudley[5], Thomas J. Walsh[6]

[1]University of Queensland Centre for Clinical Research, Brisbane, Australia; [2]University of Pittsburgh, Pittsburgh, PA, USA; [3]Rutgers Robert Wood Johnson Medical School, New Brunswick, NJ, USA; [4]The Medicines Company, Parsippany, NJ, USA; [5]The Medicines Company, San Diego, CA, USA; [6]Weill Cornell Medicine of Cornell University, New York, NY, USA

The Medicines Company
3013 Science Park Rd.
San Diego, CA 92121 USA
8 Sylvan Way
Parsippany, NJ 07054 USA

Abstract

Background

Immunocompromised patients are at high risk for mortality due to carbapenem-resistant Enterobacteriaceae (CRE). Meropenem-vaborbactam (M-V) is a novel cyclic boronic acid beta-lactamase inhibitor combination being developed for treatment of serious gram-negative infections, including CRE. This analysis reports outcomes among immunocompromised subjects in TANGO II, a randomized, open-label comparative trial with best available therapy (BAT) in subjects with complicated urinary tract infection (cUTI), acute pyelonephritis (AP), HABP/VABP, bacteremia, and cIAI, due to known or suspected CRE.

Methods

Eligible subjects were randomized 2:1 to M-V (2g/2g every 8h) or BAT for 7 to 14 days. BAT included any of the following, alone or in combination: carbapenems, aminoglycosides, polymyxin B, colistin, tigecycline, or ceftazidime-avibactam (monotherapy only). Clinical cure was defined as complete resolution of signs or symptoms such that no further antimicrobial therapy was required.

Results

Of the 50 subjects who had a baseline pathogen (m-MITT population), 19 (38.0%) were immunocompromised (4 leukemia/lymphoma, 5 medication, 10 transplant). Among the 43 subjects with a baseline CRE pathogen (mCRE-MITT), 18 (41.9%) were immunocompromised. The most common infection types among immunocompromised subjects (mCRE-MITT) were bacteremia (61.1%), cUTI/AP (16.7%), HABP/VABP (11.1%), and cIAI (11.1%). Clinical efficacy and mortality among immunocompromised in the mCRE-MITT population are shown. M-V was associated with fewer drug-related adverse events (30.8% vs. 40.0%), serious adverse events (38.5% vs. 50.0%), and renal-related adverse events (7.7% vs. 40.0%) than BAT.

Outcomes in mCRE-MITT	M-V (n=10)	BAT (n=8)	ALL (n=18)	Difference (95% CI)
Clinical Cure at End of Treatment (EOT)	6 (60%)	2 (25%)	8 (44.4%)	+35% (-13.5% to 72.8%)
Clinical Cure at Test of Cure (EOT+7 days)	7 (70%)	0 (0%)	7 (38.9%)	+70% (26.9% to 93.3%)
All-Cause Mortality Day 28	2 (20%)	3 (37.5%)	5 (27.8%)	-17.5% (-59.2% to 29.3%)

Conclusions

In immunocompromised subjects, receipt of M-V was associated with higher clinical cure rates and a lower mortality rate than BAT (m-MITT, mCRE-MITT populations). M-V is a promising treatment option for CRE in this population.

Background

- Immunocompromised patients are at increased risk for infections due to carbapenem-resistant Enterobacteriaceae (CRE) due to factors such as prolonged hospital stays, frequent use of broad-spectrum antimicrobials, and impaired host defenses.[1,2]
- Up to 33% of all CRE patients have underlying immune compromise.[3]
- Among immunocompromised patients with infections due to CRE pathogens, mortality rates range from 30% to 60%.[2,3]
- The current "best available therapy" for CRE infections involves treatment with one or more of a limited pool of antimicrobials, which have been associated with high levels of toxicity and potential for interaction with immunosuppressive medications.[4,5]
- Meropenem-vaborbactam (M-V; VABOMERE) is a new beta-lactam-beta-lactamase inhibitor combination developed for treatment of serious gram-negative infections, including CRE.
- TANGO II is a Phase 3, multi-center, randomized, open-label comparative trial designed to evaluate efficacy and safety of M-V versus best available therapy (BAT).
- Unlike most Phase 3 studies of new antimicrobials, TANGO II included subjects with underlying immunocompromise.[6-9] These subjects represent a key subpopulation of this study.

Methods

- Phase 3, multi-center, randomized, open-label study of adults with infections due to known or suspected CRE, including complicated urinary tract infection (cUTI), acute pyelonephritis (AP), hospital-acquired/ventilator-associated bacterial pneumonia (HABP/VABP), bacteremia, or complicated intra-abdominal infection (cIAI).
- Eligible subjects were randomized 2:1 to monotherapy with M-V (2g/2g every 8h via 3-h infusion) or BAT for 7-14 days (**Figure 1**).
 - o BAT included (alone or in combination): a carbapenem, aminoglycoside, polymyxin, colistin, tigecycline, or ceftazidime-avibactam (monotherapy only).

Figure 1 . Study Schema

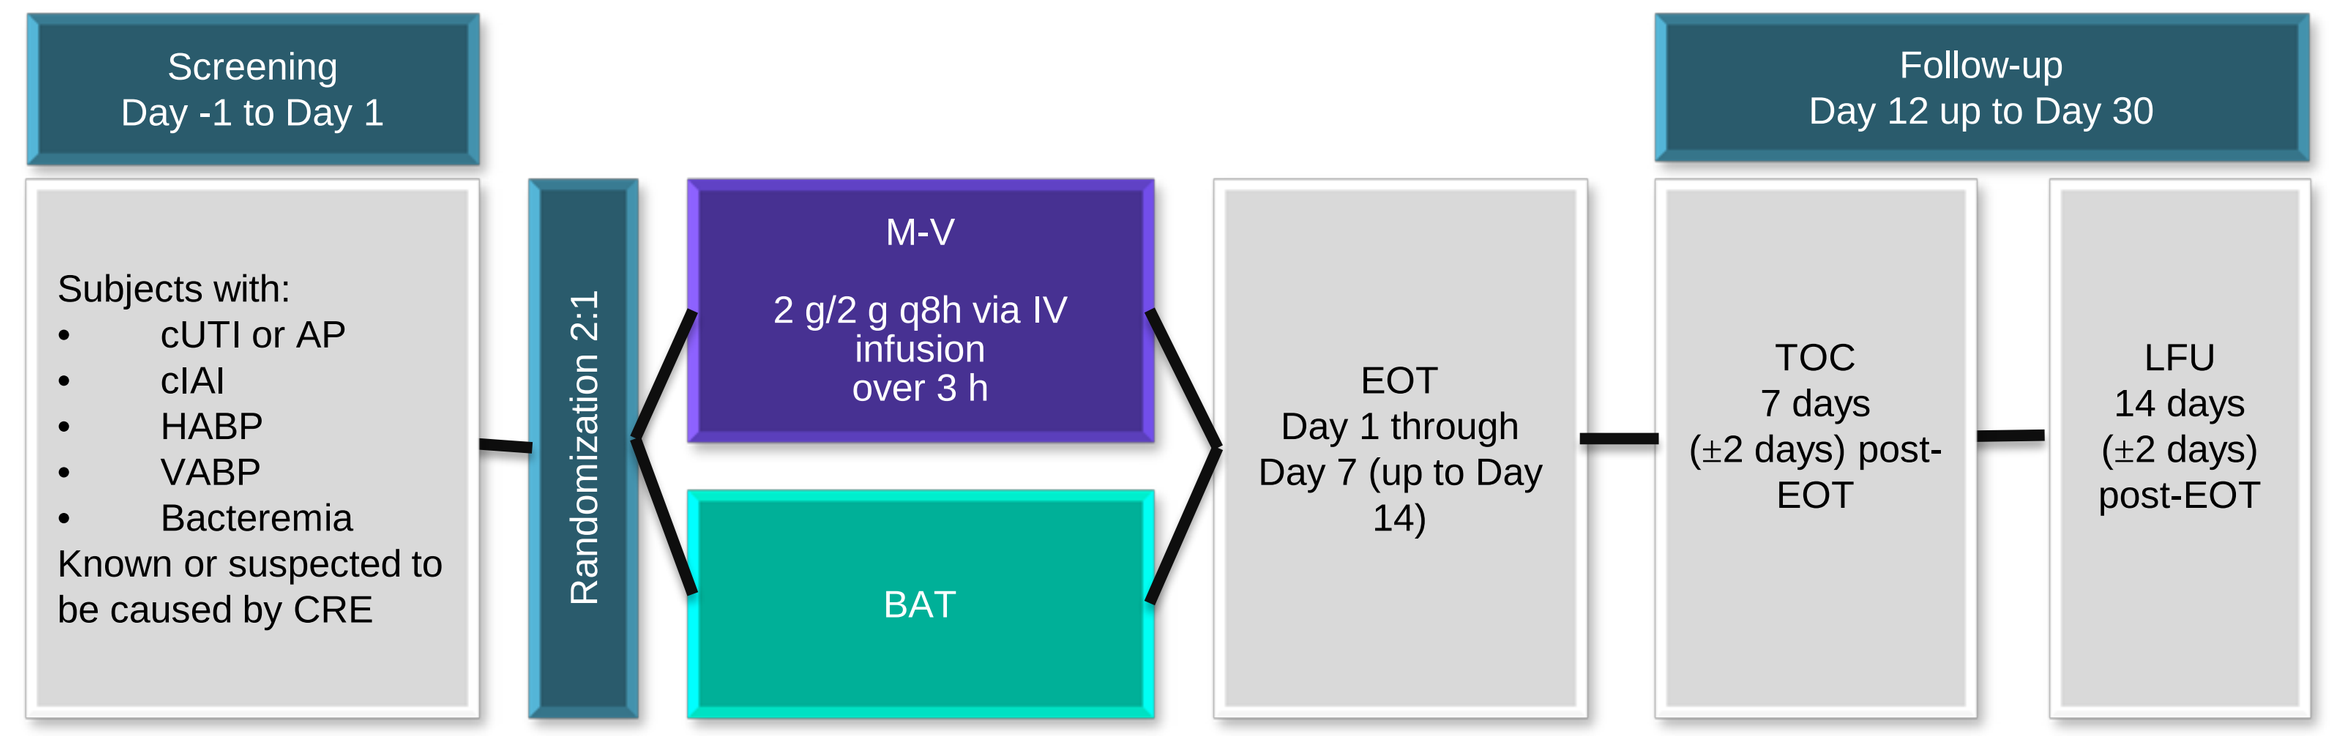
Screening
Day -1 to Day 1
Subjects with:
cUTI or AP
cIAI
HABP
VABP
Bacteremia
Known or suspected to be caused by CRE
Randomization 2:1
M-V
2 g/2 g q8h via IV infusion over 3 h
BAT
EOT
Day 1 through Day 7 (up to Day 14)
Follow-up
Day 12 up to Day 30
TOC
7 days
(±2 days) post-EOT
LFU
14 days
(±2 days)
post-EOT

EOT, end of treatment, TOC, test of cure; LFU, last follow-up.

- Enrollment was stratified by infection type and geographic region.
- Key inclusion criteria: known or suspected (evidence of CRE in culture or molecular testing within past 90 d) CRE pathogen, requirement of ≥7 days IV therapy, confirmed cUTI/AP, HABP/VABP, bacteremia, or cIAI.
- Key exclusion criteria: receipt of >24 hours of potentially effective antimicrobials (unless clinical failure), immediate life-threatening disease, known infection due to NDM, VIM, IMI or OXA-encoded beta-lactamase.
- Efforts to reduce bias included blinded investigator, blinded adjudication committee, and a source control adjudication committee (for cIAI).
- Immunocompromised was defined *a priori* as any subject with one or more of the following: medical history of active leukemia/lymphoma, history of prior organ transplant or splenectomy, active receipt of immunosuppressive medications including high-dose steroids (>20 mg/kg/day for >2 weeks of prednisone or equivalent), active receipt of bone marrow ablative chemotherapy, and/or neutropenia during the study with an absolute neutrophil count <1000 cells/mm^3.
- Clinical cure was defined as a complete resolution of signs/symptoms such that no further antimicrobial therapy was required and was assessed by the blinded investigator (BI) and primary investigator (PI) at two time points: end of treatment (EOT) and test of cure (TOC). In cases where the assessment by the BI and PI differed, clinical cure was adjudicated by the blinded adjudication committee.

Results

- Of the 50 subjects with a baseline pathogen (microbiological modified intent-to-treat population (m-MITT)), 19 (38.0%) were immunocompromised.
 - o This included 4 (21%) from leukemia/lymphoma, 5 (26.3%) from medication (primarily high-dose systemic steroids), and 10 (52.6%) from transplant (7 solid organ transplants, 3 bone marrow/stem cell transplants).
- Of the 43 subjects with a baseline CRE pathogen (microbiological CRE modified intent-to-treat population [mCRE-MITT]), 18 (41.9%) were immunocompromised. Baseline characteristics and baseline pathogens of immunocompromised subjects are shown in **Table 1**.
 - o The most common infection types among immunocompromised subjects (mCRE-MITT) were bacteremia (11/18, 61.1%), cUTI/AP (3/18, 16.7%), HABP/VABP (2/18, 11.1%), and cIAI (2/18,11.1%).

Table 1 . Baseline Characteristics of Immunocompromised Subjects (mCRE-MITT)

	M-V N=10 n, (%)	BAT N=8 n, (%)
Immunocompromising Condition		
Leukemia/lymphoma	2 (20.0)	2 (25.0)
Medications	4 (40.0)	1 (12.5)
Steroids[1]	3 (30.0)	1 (12.5)
Other[2]	1 (10.0)	0 (0.0)
Transplant	4 (40.0)	5 (62.5)
Solid Organ	2 (20.0)	5 (62.5)
Bone marrow/stem cell transplant[3]	2 (20.0)	0 (0.0)
Microbiology of baseline pathogens[4]		
Klebsiella pneumoniae	8 (80.0)	7 (87.5)
Enterobacter cloacae species complex	1 (10.0)	0 (0.0)
Enterococcus faecium	0 (0.0)	1 (12.5)
Escherichia coli	1 (10.0)	0 (0.0)
Serratia marcescens	0 (0.0)	1 (12.5)

[1] >20 mg/kg/d prednisone or equivalent.
[2] Anti-rejection medications, bone marrow suppressive chemotherapy.
[3] One bone marrow transplant and one peripheral stem cell transplant.
[4] Baseline pathogens are identified based on both local and central labs. Subjects may have had more than one baseline pathogen.

- Clinical cure rates for mCRE-MITT immunocompromised subjects at EOT and TOC (EOT+7 days) were higher for those in the M-V arm compared to the BAT arm (absolute increase 47.5%, EOT and 70%, TOC) (**Table 2**).
 - o At TOC, the increase in cure rates among immunocompromised subjects receiving M-V vs. BAT achieved statistical significance (95% CI 41.6 - 98.4, *P*<.0001) on ad hoc analysis.
- Rates of microbial cure (defined as either microbial eradication or presumed eradication) for mCRE-MITT immunocompromised subjects were higher for those in the M-V arm vs. BAT arm (absolute increase of 47.5% at EOT and TOC).
- The all-cause mortality at Day 28 was lower for those in the M-V arm vs. BAT arm (20% vs. 37.5%) for mCRE-MITT. This was associated with an absolute risk reduction (mortality) of 17.5% and relative risk reduction of 46.7%.

Table 2. Clinical and Microbial Cure Rates and All-Cause Mortality in Immunocompromised Subjects (mCRE-MITT)

	M-V N=10 n, (%)	BAT N=8 n, (%)	Absolute Difference (M-V – BAT) (95% CI)*	Relative Difference [(M-V – BAT)/ BAT]
Clinical Cure, EOT	6 (60)	1 (12.5)	47.5% (9.5 - 85.5; *P*=.01)	380%
Clinical Cure, TOC (EOT+7 days)	**7 (70)**	**0 (0)**	**70% (41.6 - 98.4; *P*<.0001)**	NA†
Microbial Cure‡, EOT	6 (60)	1 (12.5)	47.5	380%
Microbial Cure‡, TOC (EOT+7 days)	6 (60)	1 (12.5)	47.5	380%
Day 28 All-Cause Mortality	2 (20)	3 (37.5)	-17.5% (-59.2% - 24.2%; *P*=.41)	−46.7%

CI, confidence interval.
Bold font denotes values for which the 95% CI does not cross zero; therefore, statistical significance has been achieved.
* Difference estimates, 95% CI and P values were obtained by Wald test of equality.
† Approaches infinity as denominator is zero.
‡ Composite of microbial eradication or presumed eradication at respective visit.

- The maximum post baseline creatinine increase of <0.5 mg/dL and ≥0.5 mg/dL are shown in **Figure 2**.

Figure 2. Maximum Post Baseline Creatinine Increase in Immunocompromised Subjects (mCRE-MITT)

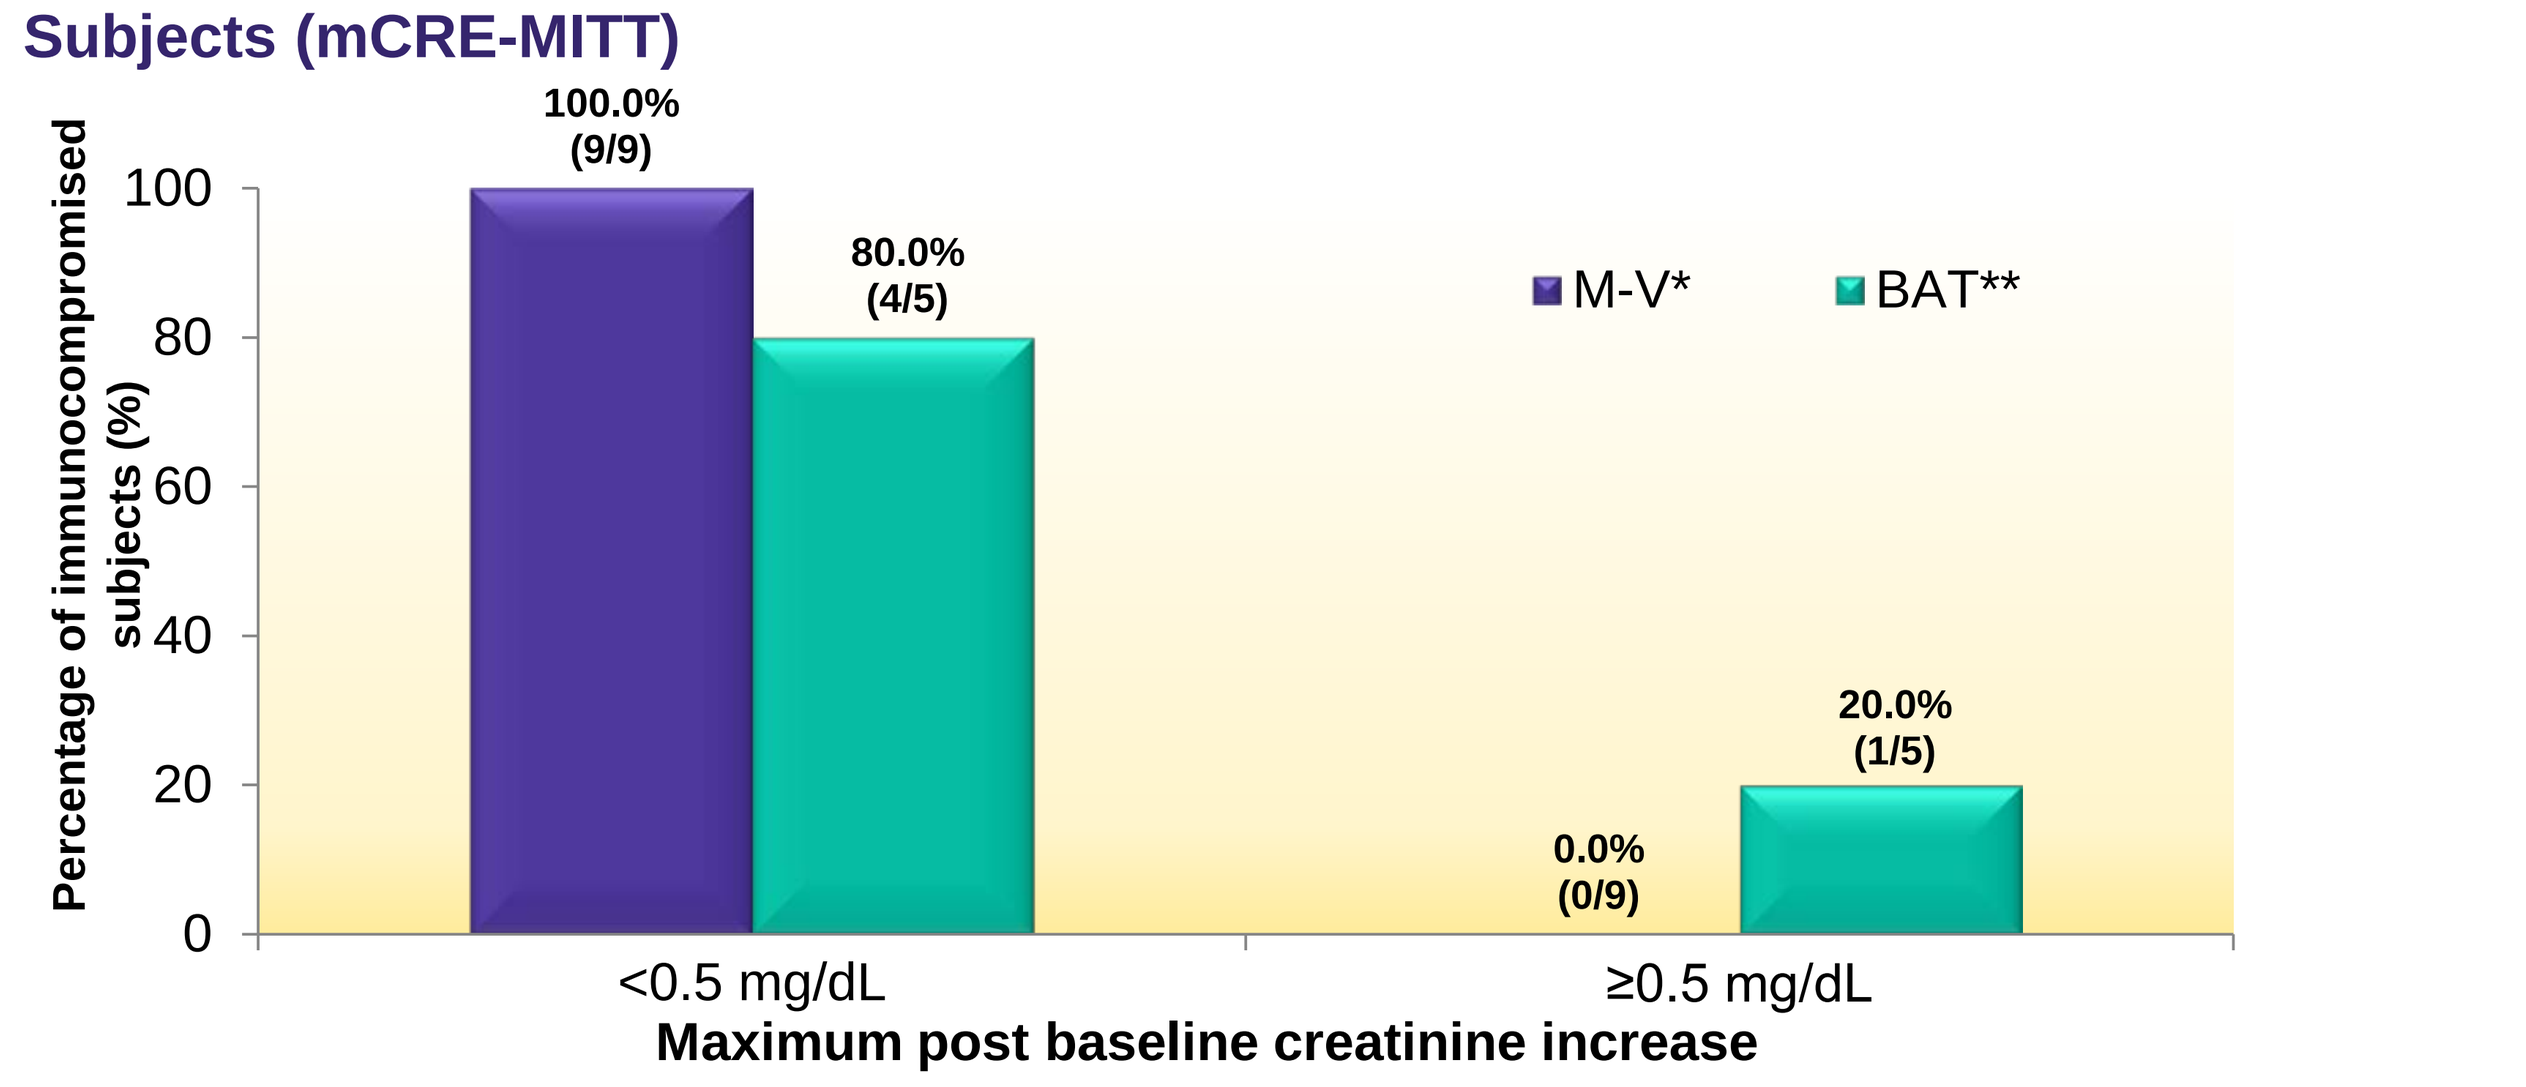
Percentage of immunocompromised subjects (%)
100
80
60
40
20
0
100.0% (9/9)
80.0% (4/5)
0.0% (0/9)
20.0% (1/5)
M-V*
BAT**
<0.5 mg/dL
≥0.5 mg/dL
Maximum post baseline creatinine increase

* 1 subject in the M-V arm was indeterminate/not assessed/censored.
** 3 subjects in the BAT arm were indeterminate/not assessed/censored.

- Among immunocompromised subjects, when compared to BAT, M-V was associated with fewer adverse events (AEs; 84.6% vs. 100%), drug-related AEs (30.8% vs. 40.0%), serious AEs (38.5% vs. 50.0%), discontinuations of study drug or study due to AEs (15.4% vs. 30.0%) and renal-related AEs (7.7% vs. 40.0%) (**Table 3**).

Table 3. Adverse Events and Safety Endpoints in Immunocompromised Subjects (Safety Population)

Adverse Events	M-V N=13 n, (%)	BAT N=10 n, (%)	Total N=23 n, (%)
TEAEs, n (%)			
Any	11 (84.6)	10 (100)	21 (91.3)
Drug-related	4 (30.8)	4 (40.0)	8 (34.8)
SAEs, n (%)			
All	5 (38.5)	5 (50.0)	10 (43.5)
Drug-related	0 (0.0)	1 (10.0)	1 (4.3)
Study drug discontinuations due to TEAEs, n (%)	2 (15.4)	3 (30.0)	5 (21.7)
Study discontinuations due to TEAEs, n (%)	2 (15.4)	3 (30.0)	5 (21.7)
Renal-related safety endpoints			
Renal-related TRAEs (Preferred Term)	1 (7.7)	4 (40.0)	5 (21.7)
Anuria	1 (7.7)	0 (0.0)	1 (4.3)
Renal failure acute	0 (0.0)	2 (20.0)	2 (8.7)
Renal impairment	0 (0.0)	2 (20.0)	2 (8.7)

SAE, serious adverse event; TEAE, treatment-emergent adverse event; TRAE, treatment-related adverse event.

Conclusions

- An immunocompromised state represents a common and important underlying comorbidity for CRE infections.
- Approximately 40% of patients with CRE in TANGO II were immunocompromised.
- Treatment of immunocompromised CRE patients with M-V was associated with higher clinical cure rate (M-V 60% vs. BAT 12.5%, *P*=.01) and lower mortality (M-V 20% vs. BAT 37%).
- Treatment with M-V in immunocompromised patients was safer than BAT as demonstrated by decreased AEs, including SAEs and renal-related AEs.
- Adverse event rates are comparable to those seen in the overall M-V population.
- M-V is a rational treatment choice for immunocompromised patients with CRE.

References

1. Pouch SM, Satlin MJ. Carbapenem-resistant Enterobacteriaceae in special populations: Solid organ transplant recipients, stem cell transplant recipients, and patients with hematologic malignancies. *Virulence.* 2017;8(4):391-402.
2. Satlin MJ, Jenkins SG, Walsh TJ. The global challenge of carbapenem-resistant Enterobacteriaceae in transplant recipients and patients with hematologic malignancies. *Clin Infect Dis.* 2014;58(9):1274-1283.
3. Alexander E, Loutit J, Tumbarello M, et al. Carbapenem-resistant Enterobacteriaceae infections: results from a retrospective series and implications for the design of prospective clinical trial. *Open Forum Infect Dis.* 2017; 4(2):ofx063.
4. Falagas ME, Lourida P, Poulikakos P, et al. Antibiotic treatment of infections due to carbapenem-resistant Enterobacteriaceae: systematic evaluation of the available evidence. *Antimicrob Agents Chemother.* 2014;58(2):654-663.
5. Munoz-Price LS, Poirel L, Bonomo R, et al. Clinical epidemiology of the global expansion of Klebsiella pneumoniae carbapenemases. *Lancet Infect Dis.* 2013;13(9):785-796.
6. Carmeli Y, Armstrong J, Laud PJ, et al. Ceftazidime-avibactam or best available therapy in patients with ceftazidime-resistant Enterobacteriaceae and Pseudomonas aeruginosa complicated urinary tract infections or complicated intra-abdominal infections (REPRISE): a randomized, pathogen-directed, phase 3 study. *Lancet Infect Dis.* 2016;16(6):661-673.
7. Mazuski JE, Gasinki LB, Armstrong J, et al Efficacy and safety of ceftazidime-avibactam plus metronidazole versus meropenem in the treatment of complicated intra-abdominal infection: results from a randomized, controlled, double-blind, phase 3 program. *Clin Infect Dis.* 2016;62(11):1380-1389.
8. Wagenlehner FM, Umeh O, Steenbergen J, et al. Ceftolazone-tazobactam compared with levofloxacin in the treatment of complicated urinary-tract infections, including pyelonephritis: a randomized, double-blind, phase 3 trial (ASPECT-cUTI). *Lancet.* 2015;385(9981):1949-1956.
9. National Institutes of Health. Study of the safety, tolerability and efficacy of MK-7655+imipenem+cilastin versus imipenem/cilastin alone for the treatment of complicated urinary tract infection (MK-7655-003). Available at: https://clinicaltrials.gov/ct2/show/NCT01505634. NLM Identifier: NCT01505634.

Disclosures

This project has been funded in whole or in part with federal funds from the Department of Health and Human Services; Office of the Assistant Secretary for Preparedness and Response; Biomedical Advanced Research and Development Authority (BARDA), under Contract No. HHSO100201400002C with Rempex Pharmaceuticals, a wholly-owned subsidiary of The Medicines Company and Agreement No. HHSO100201600026C with The Medicines Company.

Presented at IDWeek 2017, October 4-8, 2017, San Diego, CA