ECCMID 2019 | Poster #P1882

Oritavancin Activity against *Staphylococcus aureus* Clinical Isolates Causing Serious Infections in Hospitalized Patients in Europe (2017–2018)

Cecilia G. Carvalhaes[1], Helio S. Sader[1], Jennifer M. Streit[1], Robert K. Flamm[1], Rodrigo E. Mendes[1]
[1] JMI Laboratories, North Liberty, Iowa, USA

Introduction

- Oritavancin, a potent lipoglycopeptide antibiotic, with a convenient single dose regimen, is a desirable option for serious Gram-positive infections as well as for those requiring prolonged treatment courses
- Oritavancin has potent bactericidal activity by inhibiting peptidoglycan synthesis at 2 stages and by causing depolarization of the bacterial cell membrane
- The European Medicines Agency and the United States Food and Drug Administration approved oritavancin for the treatment of skin and soft tissue infections in 2015
- This study assessed the activity of oritavancin and comparators against a contemporary collection of *Staphylococcus aureus* isolates causing serious infections in Europe

Materials and Methods

Organism collection

- A total of 1,232 *S. aureus* isolates were included that were deemed to be the primary organisms responsible for invasive infections during 2017–2018 in 33 medical centres in 13 European and adjacent countries/regions
- The list of participating countries, sites, and number of isolates is shown in Table 1
- Only 1 isolate per patient infection episode was included
- Bacterial identification was confirmed by matrix-assisted laser desorption ionization–time of flight mass spectrometry (Bruker Daltonics, Massachusetts, USA) following the manufacturer's instructions

Antimicrobial susceptibility testing

- Susceptibility testing was performed by broth microdilution according to European Committee of Antimicrobial Susceptibility Testing (EUCAST) methods and determinations were based on EUCAST (2019) breakpoint criteria
- Oritavancin minimal inhibitory concentrations (MICs) were determined in the presence of polysorbate-80 (0.002%)
- Bacterial inoculum density was monitored by colony counts to ensure an adequate number of cells for each testing event
- Quality assurance was performed by concurrently testing EUCAST-recommended quality control (QC) reference strains (*S. aureus* ATCC 29213 and *Enterococcus faecalis* ATCC 29212), and all results were within published acceptable ranges

Figure 1 Distribution of *S. aureus* isolates by infection type

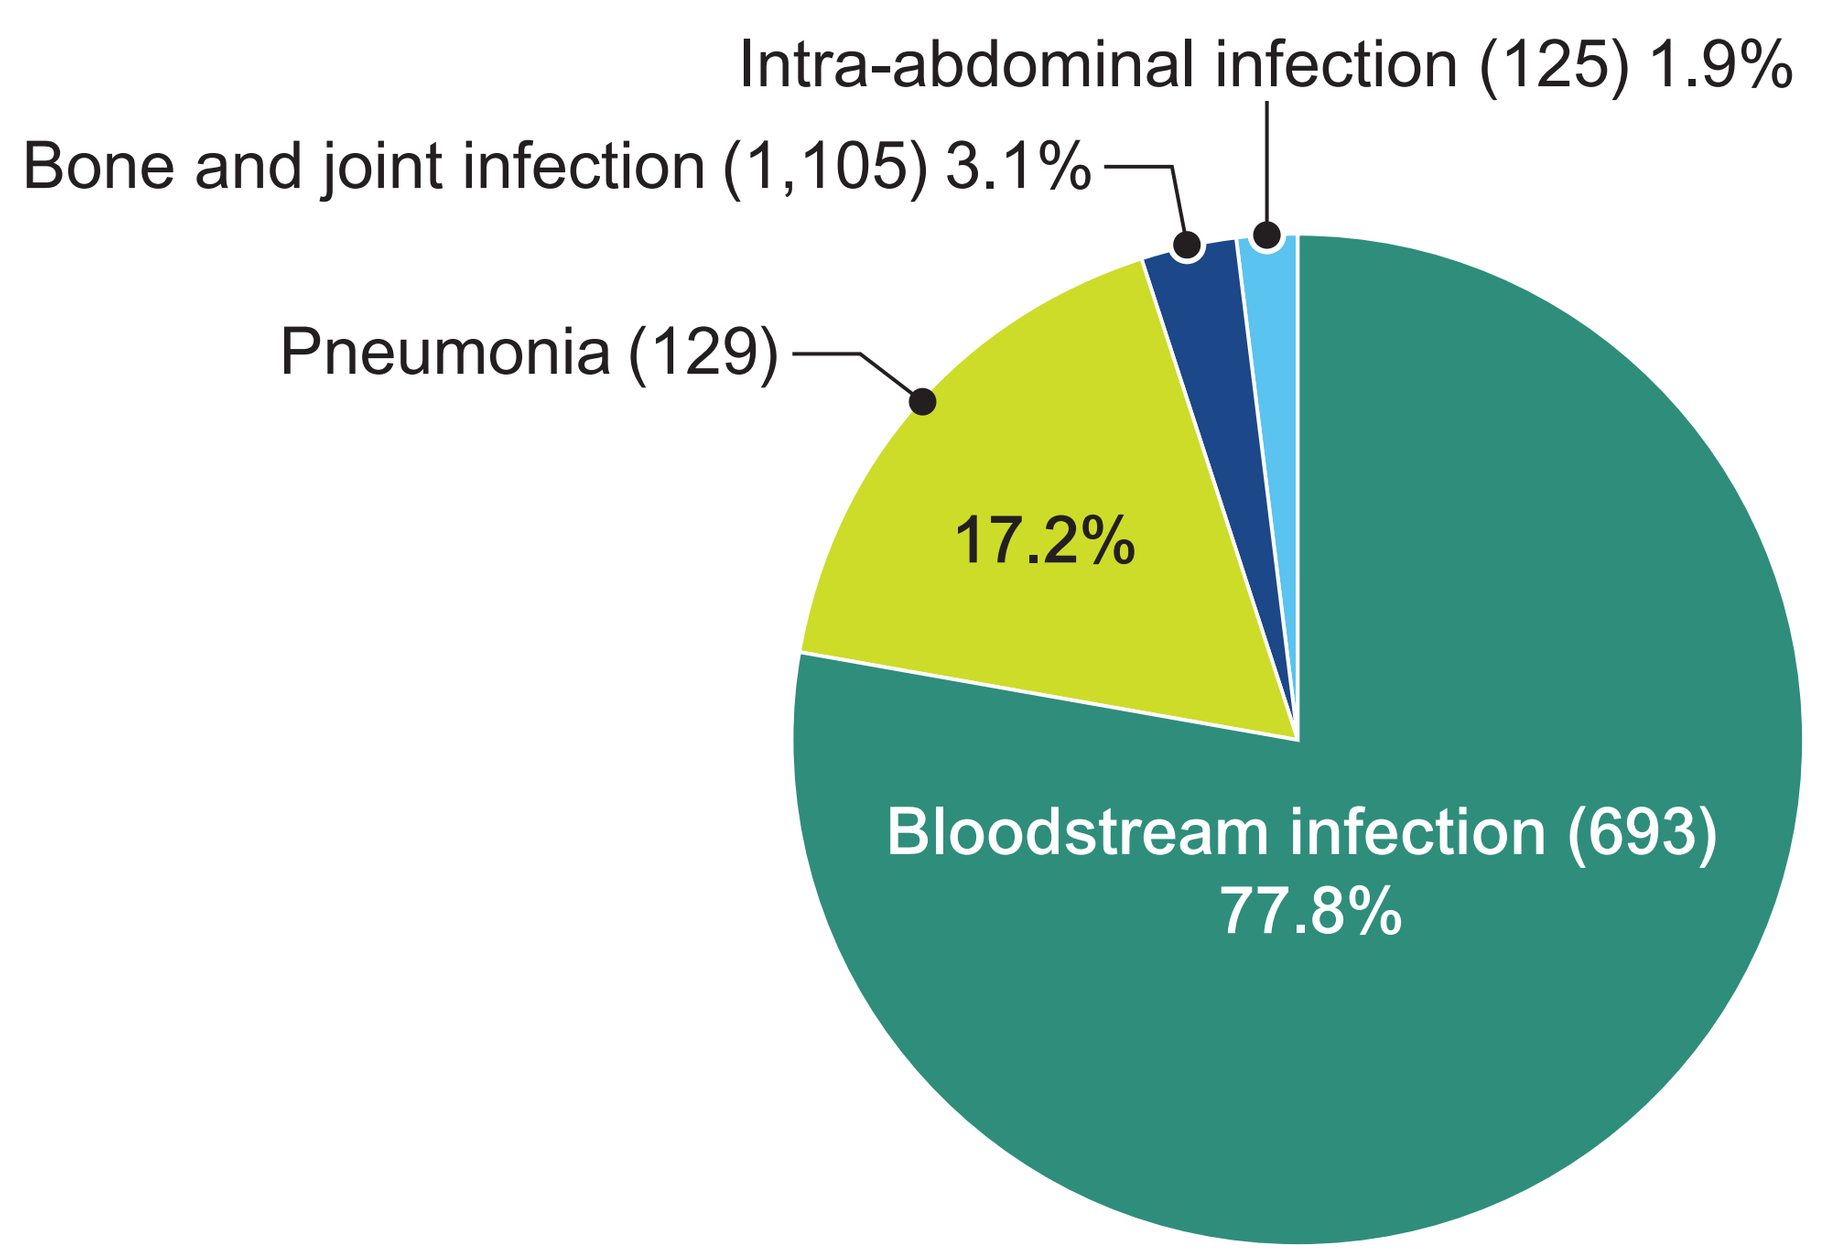
Intra-abdominal infection (125) 1.9%
Bone and joint infection (1,105) 3.1%
Pneumonia (129)
17.2%
Bloodstream infection (693)
77.8%

Results

- *S. aureus* was most frequently recovered from bloodstream infections (BSIs; 77.8% of all isolates), followed by pneumonia (17.2%), bone and joint infections (BJIs; 3.1%), and intra-abdominal infections (IAIs; 1.9%, Figure 1)
- Oritavancin (MIC_{50}/MIC_{90}, 0.03/0.06 mg/L) inhibited all *S. aureus* isolates at the susceptible breakpoint of ≤0.12 mg/L (Table 2)
- Testing against methicillin-resistant *S. aureus* (MRSA; 23.3% of all *S. aureus*) and methicillin-susceptible *S. aureus* (MSSA) groups yielded similar oritavancin MIC_{50} (0.03 mg/L) and MIC_{90} (0.03-0.06 mg/L) results, regardless of infection type (Table 4)
- MRSA rates varied greatly among European countries (0%-46.6%; Table 3), but oritavancin MIC_{50} and MIC_{90} values remained at 0.03 mg/L and 0.03-0.06 mg/L, respectively (data not shown)
- MSSA isolates showed high overall susceptibility rates to oritavancin (100%), vancomycin (100%), teicoplanin (99.9%), linezolid (100%), and clindamycin (98.6%), regardless of infection type
- Oritavancin, vancomycin, teicoplanin, and linezolid remained active against MRSA isolates, regardless of infection type (Figure 2)
 – Oritavancin had MIC_{50}/MIC_{90} results (0.03/0.06 mg/L) 16- to 32-fold lower than vancomycin (MIC_{50}/MIC_{90}, 1 / 1 mg/L), teicoplanin (MIC_{50}/MIC_{90}, 0.5 / 1 mg/L), and linezolid (MIC_{50}/MIC_{90}, 1 / 2 mg/L, Table 4)
- Decreased susceptibility to vancomycin (MIC, 2 mg/L) was observed in 8 MRSA isolates causing bloodstream infection, which exhibited oritavancin MIC values of 0.03 mg/L (4 isolates), 0.06 mg/L (3 isolates), and 0.12 mg/L (1 isolate)
- Three teicoplanin-resistant *S. aureus* isolates (MIC, >2 mg/L) were observed; 2 of them also showed reduced susceptibility of vancomycin (MIC, 2 mg/L) and showed oritavancin MIC values of 0.03-0.12 mg/L

Table 1 List of European countries and number of *S. aureus* isolates causing serious infections

Country	No. of sites	No. of isolates
Belgium	1	9
France	5	238
Germany	5	144
Greece	1	12
Ireland	2	61
Italy	4	311
Poland	1	1
Portugal	1	47
Russia	3	72
Spain	3	90
Sweden	2	87
Turkey	2	69
United Kingdom	3	91
Total	33	1,232

Table 2 MIC distribution of oritavancin and comparator agents against 1,232 *S. aureus* isolates

Antimicrobial agent	Antimicrobial range (mg/L)	No. and cumulative % of isolates inhibited at MIC (mg/L) of:													MIC_{50}	MIC_{90}
		0.008	0.015	0.03	0.06	0.12	0.25	0.5	1	2	4	8	16	>[a]	(mg/L)	
Oritavancin	0.004 - 1	17	344	747	99	25									0.03	0.06
		1.4%	29.3%	89.9%	98.0%	100.0%										
Clindamycin	≤0.03 - >2			290	830	41	5	2	0	0				64	0.06	0.06
				23.5%	90.9%	94.2%	94.6%	94.8%	94.8%	94.8%				100.0%		
Levofloxacin	≤0.03 - >4			1	5	224	679	48	10	3	13			249	0.25	>4
				0.1%	0.5%	18.7%	73.8%	77.7%	78.5%	78.7%	79.8%			100.0%		
Linezolid	≤0.12 - >8						0	54	798	375	5				1	2
							0.0%	4.4%	69.2%	99.6%	100.0%					
Teicoplanin	≤0.12 - 16					6	282	857	71	13	2	1			0.5	0.5
						0.5%	23.4%	92.9%	98.7%	99.8%	99.9%	100.0%				
Vancomycin	≤0.12 - 4					0	4	255	952	20					1	1
						0.0%	0.3%	21.0%	98.4%	100.0%						

[a] Greater than the highest concentration tested.

Figure 2 Oritavancin and comparators activity against MRSA isolates

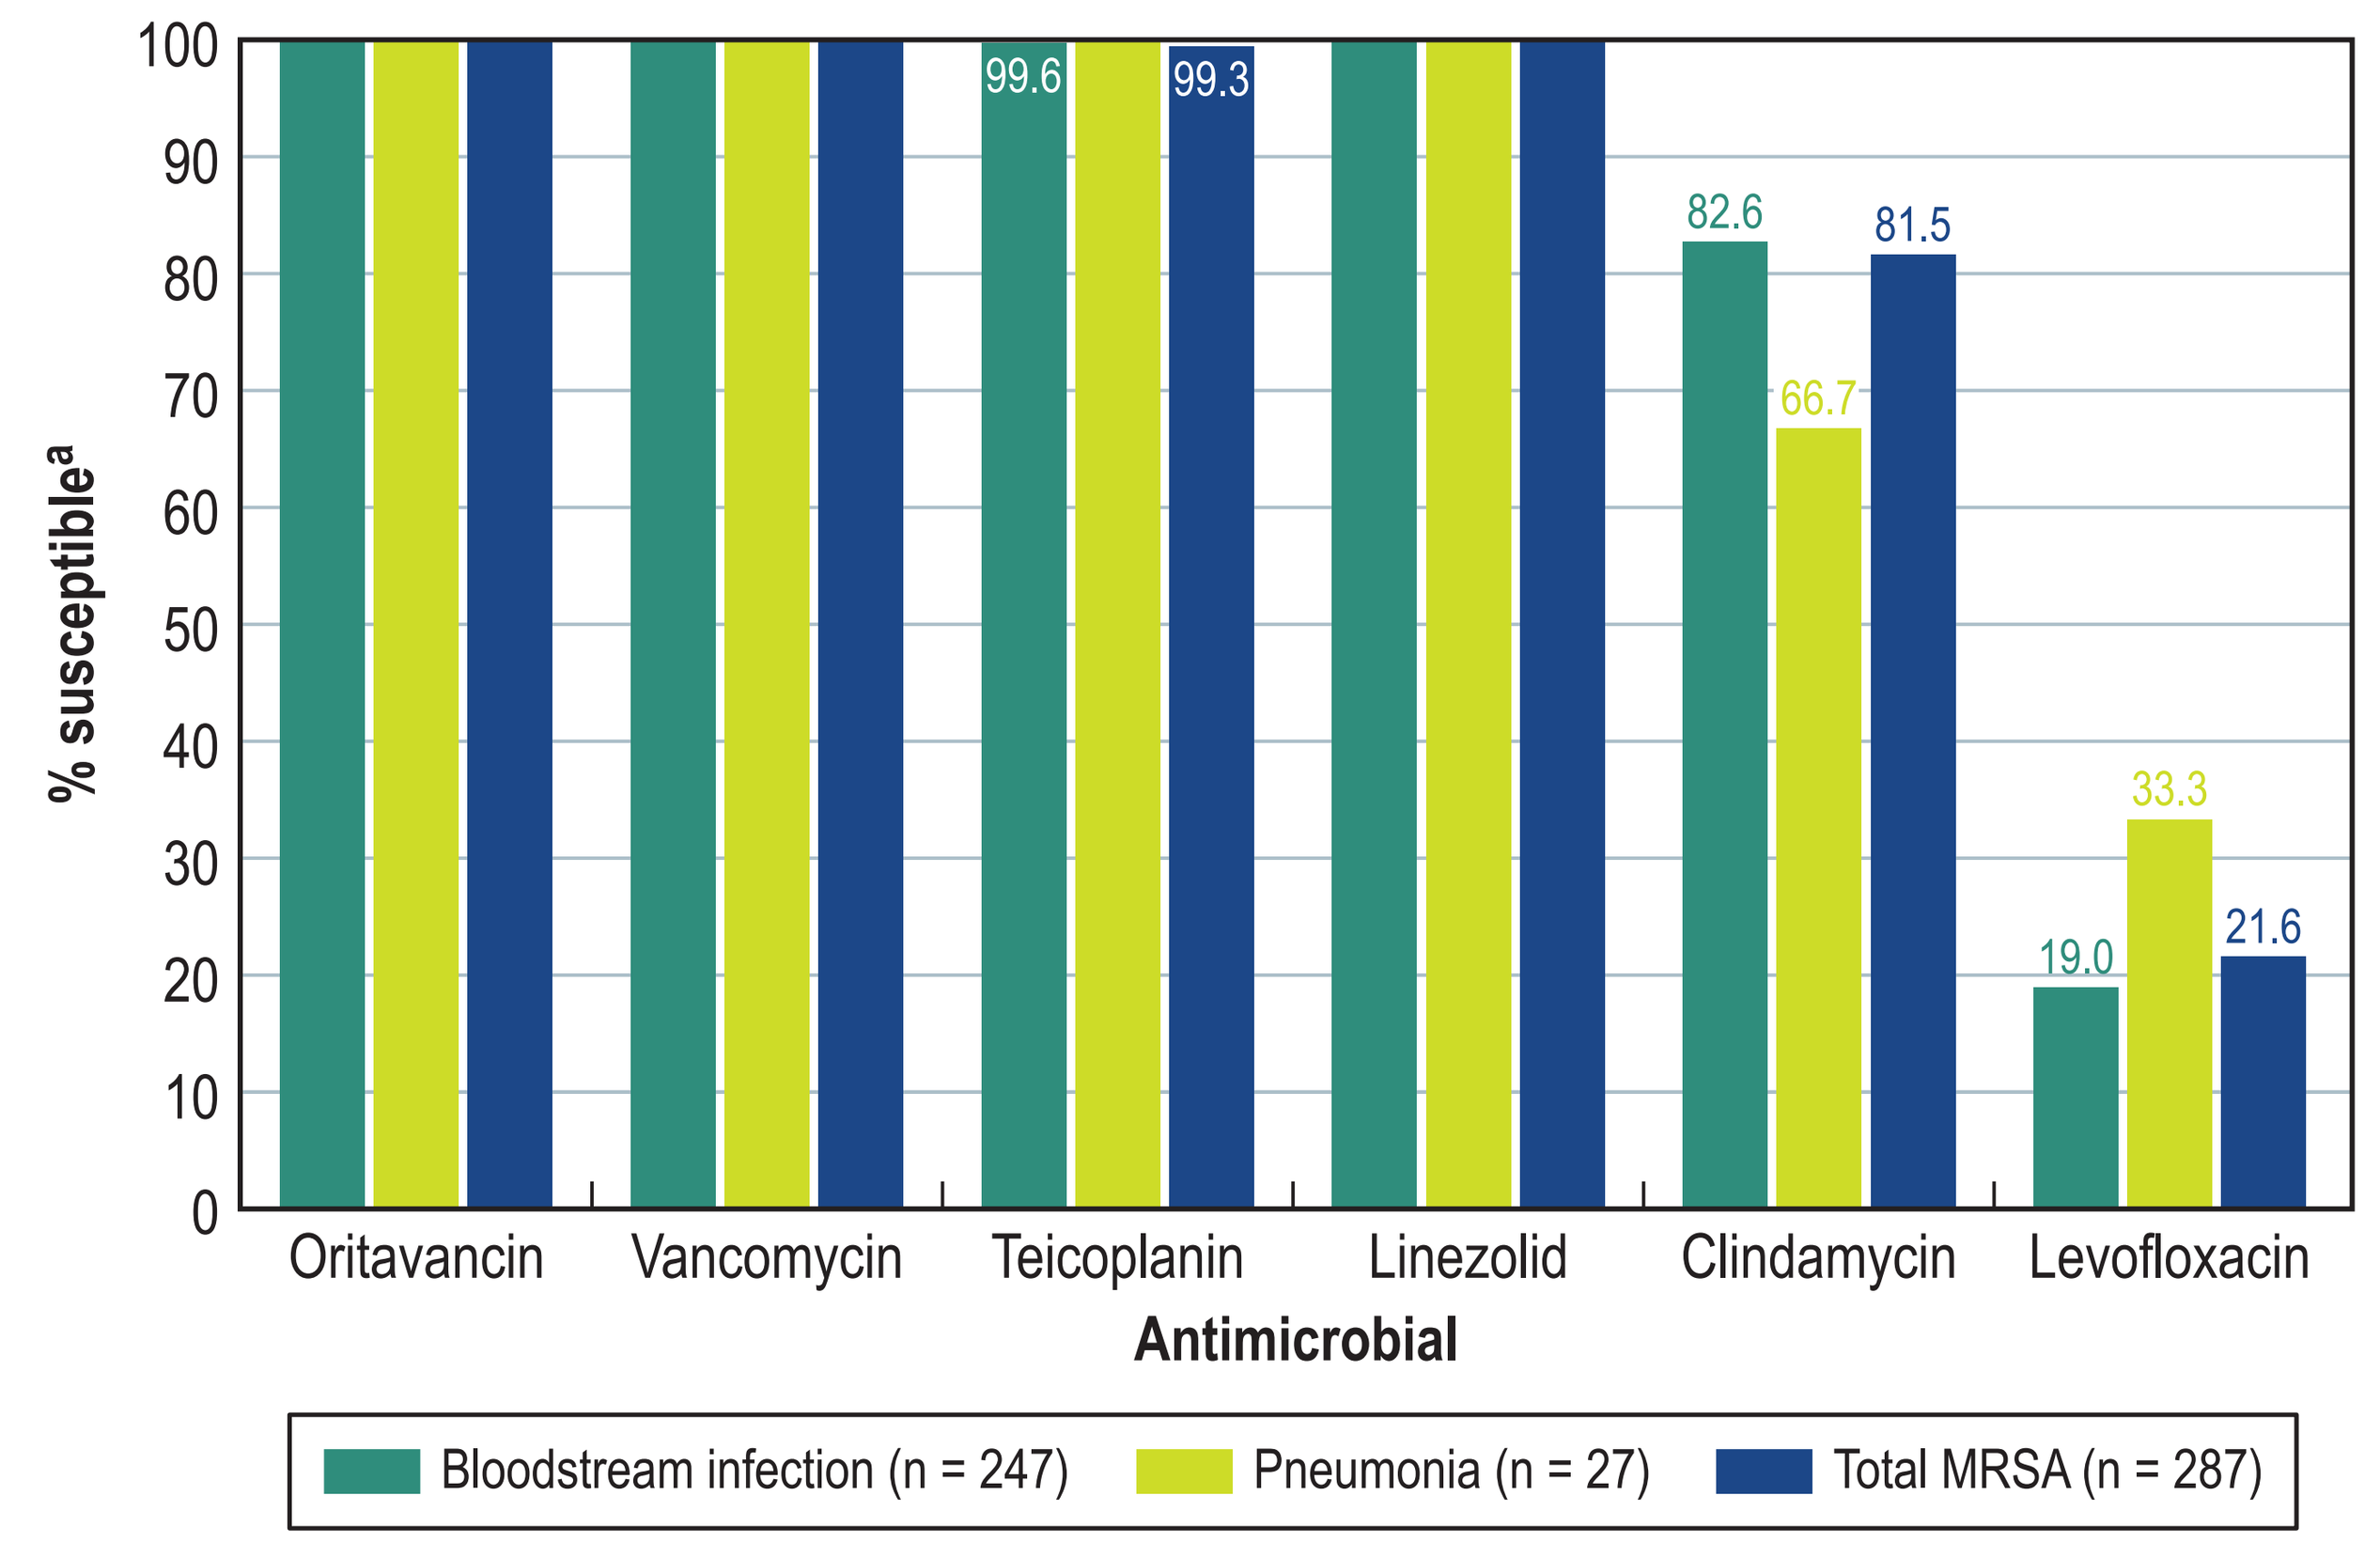
100
90
80
70
60
50
40
30
20
10
0
% susceptible[a]
99.6
99.3
82.6
81.5
66.7
33.3
19.0
21.6
Oritavancin
Vancomycin
Teicoplanin
Linezolid
Clindamycin
Levofloxacin
Antimicrobial
Bloodstream infection (n = 247)
Pneumonia (n = 27)
Total MRSA (n = 287)

[a] Susceptibility rate based on EUCAST v.9.0 criteria.

Table 3 MRSA rates among European countries and surroundings

Country	Total isolates	% MRSA	TZD MIC_{50} (mg/L)	TZD MIC_{90} (mg/L)
Italy	311	46.60%	0.03	0.03
Turkey	69	27.50%	0.03	0.06
Spain	90	25.60%	0.03	0.03
Germany	144	19.40%	0.03	0.06
Portugal	47	19.10%	0.03	0.03
Russia	72	18.10%	0.03	0.06
Ireland	61	16.40%	0.03	0.06
France	238	12.20%	0.03	0.03
United Kingdom	91	9.90%	0.03	0.03
Greece	12	8.30%	0.03	0.06
Sweden	87	1.10%	0.03	0.06
Belgium	9	0.00%	0.03	0.06
Poland	1	0.00%	0.03	—
Total	1232	23.30%	0.03	0.06

Table 4 Activity of oritavancin and comparators against *S. aureus* isolates by infection type in Europe

Infection type	$MIC_{50/90}$ (mg/L)					
(no. of isolates)	Oritavancin	Vancomycin	Teicoplanin	Linezolid	Clindamycin	Levofloxacin
MSSA (945)	0.03 / 0.03	1 / 1	0.5 / 0.5	1 / 2	0.06 / 0.06	0.25 / 0.25
BSI (712)	0.03 / 0.03	1 / 1	0.5 / 0.5	1 / 2	0.06 / 0.06	0.25 / 0.25
Pneumonia (185)	0.03 / 0.06	1 / 1	0.5 / 0.5	1 / 2	0.06 / 0.06	0.25 / 0.25
IAI (17)	0.03 / 0.06	1 / 1	0.5 / 0.5	1 / 2	0.06 / >2	0.25 / 0.5
BJI (31)	0.03 / 0.03	1 / 1	0.5 / 0.5	1 / 2	0.06 / 0.06	0.25 / 0.25
MRSA (287)	0.03 / 0.06	1 / 1	0.5 / 1	1 / 2	0.06 / >2	>4 / >4
BSI (247)	0.03 / 0.06	1 / 1	0.5 / 1	1 / 2	0.06 / >2	>4 / >4
Pneumonia (27)	0.03 / 0.06	1 / 1	0.5 / 1	1 / 2	0.06 / >2	>4 / >4
IAI (6)	0.03 / -	1 / -	0.5 / -	1 / -	0.06 / -	0.25 / -
BJI (7)	0.03 / -	1 / -	0.5 / -	1 / -	0.06 / -	4 / -

[a] Susceptibility rate based on EUCAST v.9.0 criteria.
[b] MSSA, methicillin-susceptible *S. aureus*; MRSA, methicillin-resistant *S. aureus*; BSI, bloodstream infection; IAI, intra-abdominal infection; BJI, bone and joint infection.

Conclusions

- Oritavancin showed good activity (MIC_{50}/MIC_{90}, 0.03/0.06 mg/L) against this 2017–2018 European collection of *S. aureus* isolates responsible for pneumonia, intra-abdominal, bone and joint, and bloodstream infections
- All isolates were susceptible to oritavancin, vancomycin, and linezolid, regardless of infection type or methicillin resistance
- Oritavancin showed 16- to 32-fold higher activity than vancomycin, teicoplanin, and linezolid against *S. aureus* isolates causing serious infections
- Oritavancin was also active against MRSA isolates displaying reduced susceptibility to vancomycin and/or teicoplanin
- A prolonged half-life and high potency may support oritavancin as a good option for treating serious *S. aureus* infections

Acknowledgements

Funding for this research was provided by Melinta Therapeutics, Morristown, NJ, USA.

References

Clinical and Laboratory Standards Institute (2019). *M100Ed29E. Performance standards for antimicrobial susceptibility testing: 29th informational supplement.* Wayne, PA: CLSI.

EUCAST (2019). Breakpoint tables for interpretation of MIC's and zone diameters. Version 9.0, January 2019. Available at: http://www.eucast.org/fileadmin/src/media/PDFs/EUCAST_files/Breakpoint_tables/v_9.0_Breakpoint_Tables.pdf. Accessed January 2019.

Lowy FD. *Staphylococcus aureus* infections. N Engl J Med 1998;339:520–32.

European Centre for Disease Prevention and Control. Point prevalence survey of healthcareassociated infections and antimicrobial use in European acute care hospitals. Stockholm: ECDC; 2013.

van Hal SJ, Jensen SO, Vaska VL, Espedido BA, Paterson DL, Gosbell IB. Predictors of mortality in *Staphylococcus aureus* bacteremia. Clin Microbiol Rev. 2012; 25:362–86.

Contact

Rodrigo E. Mendes, PhD
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: rodrigo-mendes@jmilabs.com

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/ECCMID19-oritavancin-staphylococci.pdf

Charges may apply. No personal information is stored.

ECCMID 2019, April 13–16, 2019, Amsterdam, the Netherlands