IDWEEK 2020 | Poster number: 1620

Minocycline Activity Against Unusual Clinically Significant Gram-Negative Pathogens

Dee Shortridge, S.J. Ryan Arends, Jennifer M. Streit, Mariana Castanheira
JMI Laboratories, North Liberty, IA, USA

Introduction

- Unusual non-glucose fermenting Gram-negative (NFGN) pathogens, including *Achromobacter* spp., *Alcaligenes* spp., *Aeromonas* spp., and other genera, can cause serious hospital-acquired infections in immunocompromised patients.
 - Some genera are inherently resistant to common drug classes, and can acquire other resistance mechanisms, making them difficult to treat.
 - Publications with susceptibilities of NFGN isolates are uncommon.
- In this study, we analyzed the susceptibility of unusual NFGN isolates to minocycline. Isolates were collected as part of the SENTRY Antimicrobial Surveillance Program from 2014–2019.
 - The most common NFGN species (*Pseudomonas aeruginosa, Acinetobacter baumannii-calcoaceticus* species complex, and *Stenotrophomonas maltophilia*) were excluded from this analysis.

Materials and Methods

- From 2014–2019, 1,813 unusual NFGN isolates were collected from hospitalized patients in 102 hospitals in 35 countries on 4 continents. Hospitals submitted 1 isolate per patient per infection episode that met local criteria for being the likely causative pathogen.
- Identification was performed by the submitting laboratory and confirmed by JMI Laboratories with matrix-assisted laser desorption ionization-time of flight mass spectrometry or other molecular methods if needed.
- The most common NFGN species (*Pseudomonas aeruginosa, Acinetobacter baumannii-calcoaceticus* species complex, and *Stenotrophomonas maltophilia*) were excluded from this analysis.
 - Each genus/species analyzed had at least 10 isolates.
 - Some genera had a large number of species, each with a small number of isolates. In these cases, isolates were combined for analysis. If a single species was the main species within a genus, that species is shown.
- Isolates were tested for minocycline susceptibility using the CLSI broth microdilution method at JMI Laboratories (CLSI, 2018).
 - CLSI breakpoints from M100 or M45 were used as appropriate.
 - If breakpoints were not available, *Acinetobacter* minocycline CLSI breakpoints (≤4/8/≥16 mg/L) were applied.
- All infection types were included in the susceptibility analysis.

Results

- Pneumonia in hospitalized patients was the most common infection from which the NFGN were isolated (Figure 1).
- The top 5 unusual NFGN species were: *Achromobacter* species (n=411), including *A. xylosoxidans* (n=202); *Burkholderia* spp. (n=246), including *B. cepacia* species complex (n=229); *Aeromonas* spp. (n=127), including *Aeromonas hydrophila* (n=35); *Chryseobacterium* spp. (n=59); and *Alcaligenes faecalis* (n=42).
- The MIC frequency distribution and $MIC_{50/90}$ values of minocycline for these species are shown in Table 1.
- Minocycline susceptibility was >90% for all organisms shown, except *Alcaligenes faecalis, Burkholderia* spp., and *non-aeruginosa Pseudomonas* spp.
 - Minocycline susceptibility of *A. faecalis* was 88.1%.
 - Susceptibility of *Burkholderia* spp. was 87.0% and *B. cepacia* species complex was 86.5%.
 - Susceptibility of *non-aeruginosa Pseudomonas* spp. was 89.7%.
- The susceptibilities of minocycline and other comparators are shown for the 2 largest groups, *non-baumannii-calcoaceticus Acinetobacter* spp. (Table 2) and *Achromobacter* spp. (Table 3).
 - For *Acinetobacter* spp., minocycline had the highest susceptibility (98.8%) of the comparators tested. The agent with the lowest susceptibility was ceftazidime (79.5%).
 - For *Achromobacter* spp., minocycline had the highest susceptibility, 92.7%. The agent with the lowest susceptibility was aztreonam (1.0%).

Conclusions

- Minocycline had >85% susceptibility for the most frequently isolated unusual NFGN, including 99.2% for *Aeromonas* spp., 98.8% for *non-baumannii Acinetobacter*, and 92.7% for *Achromobacter* spp.
- These data suggest that minocycline remains a useful treatment option for infections caused by unusual NFGN.

Acknowledgements

This study was supported by Melinta Therapeutics, Inc.

References

CLSI (2018). M07 Eleventh edition. Methods for Dilution Antimicrobial Susceptibility Testing for Bacteria That Grow Aerobically. Wayne, PA, Clinical and Laboratory Standards Institute.

CLSI (2020). M100 30th edition. Performance Standards for Antimicrobial Susceptibility Testing. Wayne, PA, Clinical and Laboratory Standards Institute.

CLSI (2015). M45 3rd edition. Methods for Antimicrobial Dilution and Disk Susceptibility Testing of Infrequently Isolated or Fastidious Bacteria. Wayne, PA, Clinical and Laboratory Standards Institute.

EUCAST (2020). "The European Committee on Antimicrobial Susceptibility Testing. Breakpoint tables for interpretation of MICs and zone diameters." Accessed January 2020.

Contact

Dee Shortridge, PhD
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, Iowa 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: dee-shortridge@jmilabs.com

Table 1 MIC distribution of minocycline tested against various unusual non-fermentative Gram-negative species with at least 10 isolates

Organism (number)	Dilution (mg/L) 0.03	0.06	0.12	0.25	0.5	1	2	4	8	16	32	>	MIC_{50}	MIC_{90}
Acinetobacter non-baumannii complex spp. (424)		166	131	80	32	9	0	1	1			4	0.12	0.5
		39.2%	70.0%	88.9%	96.5%	98.6%	98.6%	98.8%	99.1%			100.0%		
Achromobacter spp. (411)		2	7	21	62	141	97	51	22			8	1	4
		0.5%	2.2%	7.3%	22.4%	56.7%	80.3%	92.7%	98.1%			100.0%		
A. xylosoxidans (202)		1	3	6	36	73	35	32	10			6	1	4
		0.5%	2.0%	5.0%	22.8%	58.9%	76.2%	92.1%	97.0%			100.0%		
Aeromonas spp. (127)		0	2	10	60	35	16	3	0	0	0	1	0.5	2
		0.0%	1.6%	9.4%	56.7%	84.3%	96.9%	99.2%	99.2%	99.2%	99.2%	100.0%		
A. hydrophila (35)		0	1	4	17	10	2	1					0.5	1
		0.0%	2.9%	14.3%	62.9%	91.4%	97.1%	100.0%						
Alcaligenes faecalis (42)			0	1	1	8	19	8	3			2	2	8
			0.0%	2.4%	4.8%	23.8%	69.0%	88.1%	95.2%			100.0%		
Burkholderia spp. (246)		1	1	2	14	57	91	48	12			20	2	8
		0.4%	0.8%	1.6%	7.3%	30.5%	67.5%	87.0%	91.9%			100.0%		
B. cepacia complex (229)		1	0	2	13	51	84	47	11			20	2	8
		0.4%	0.4%	1.3%	7.0%	29.3%	65.9%	86.5%	91.3%			100.0%		
Burkholderia non-cepacia spp. (47) [a]		0	1	0	4	14	17	7	1			3	2	4
		0.0%	2.1%	2.1%	10.6%	40.4%	76.6%	91.5%	93.6%			100.0%		
Chryseobacterium spp. (59)		0	1	0	6	14	21	14	3				2	4
		0.0%	1.7%	1.7%	11.9%	35.6%	71.2%	94.9%	100.0%					
Delftia spp. (13)	0	1	2	5	5								0.25	0.5
	0.0%	7.7%	23.1%	61.5%	100.0%									
Elizabethkingia spp. (23)			0	1	12	8	1	0	1				0.5	1
			0.0%	4.3%	56.5%	91.3%	95.7%	95.7%	100.0%					
Ochrobactrum spp. (16)		0	1	1	8	6							0.5	1
		0.0%	6.2%	12.5%	62.5%	100.0%								
Pseudomonas non-aeruginosa spp. (340)		0	1	9	24	76	119	76	20			15	2	8
		0.0%	0.3%	2.9%	10.0%	32.4%	67.4%	89.7%	95.6%			100.0%		
Rhizobium spp. (15)		3	6	5	0	1							0.12	0.25
		20.0%	60.0%	93.3%	93.3%	100.0%								
Shewanella spp. (14)		0	1	6	6	1							0.25	0.5
		0.0%	7.1%	50.0%	92.9%	100.0%								
Sphinogomonas spp. (19)		13	4	0	1	1							≤0.06	0.5
		68.4%	89.5%	89.5%	94.7%	100.0%								
Stenotrophomonas non-maltophilia spp. (12)		2	6	1	3								0.12	0.5
		16.7%	66.7%	75.0%	100.0%									

CLSI minocycline breakpoints are shown by green shading for susceptible, yellow for intermediate, and orange for resistant (CLSI M100 or M45). *Acinetobacter* spp. breakpoints were used for species without breakpoints.
[a] Organisms included: *Burkholderia cenocepacia* (11), *B. gladioli* (17), *B. multivorans* (19)

Table 2 Activity of minocycline and comparator antimicrobial agents tested against 424 *Acinetobacter* isolates (excluding *Acinetobacter baumannii-calcoaceticus* species complex)

Antimicrobial agent	No. of isolates	mg/L MIC_{50}	MIC_{90}	MIC range	CLSI[a] %S	%I	%R	EUCAST[a] %S	%I	%R
Minocycline	424	0.12	0.5	≤0.06 to >8	98.8	0.2	0.9			
Meropenem-vaborbactam	424	0.25	1	≤0.015 to >32						
Meropenem	424	0.25	1	≤0.015 to >32	95.0	0.5	4.5	95.0	0.9	4.0
Imipenem	423	≤0.12	0.5	≤0.12 to >8	95.3	0.2	4.5	95.3	0.2	4.5
Tetracycline	358	2	4	≤0.5 to >8	94.1	2.2	3.6			
Amikacin	423	1	4	≤0.25 to >32	95.3	1.7	3.1	92.4 [b]		7.6
Aztreonam	424	>16	>16	0.5 to >16						
Ceftazidime	424	4	>16	≤0.25 to >16	79.5	8.3	12.3			
Colistin	423	≤0.5	2	≤0.5 to >8		90.5	9.5	90.5		9.5
Gentamicin	424	≤1	2	≤1 to >8	94.3	1.7	4.0	94.3 [b]		5.7
Levofloxacin	424	≤0.12	0.5	≤0.12 to >4	97.2	0.9	1.9	95.0	1.4	3.5

[a] Criteria as published by CLSI (2020) and EUCAST (2020).
[b] For infections originating from the urinary tract. For systemic infections, aminoglycosides must be used in combination with other active therapy.
Organisms include: *Acinetobacter beijerinckii* (3), *A. berezinae* (34), *A. calcoaceticus* (2), *A. courvalinii* (6), *A. guillouiae* (8), *A. haemolyticus* (14), *A. johnsonii* (20), *A. junii* (27), *A. lwoffii* (78), *A. nosocomialis* (8), *A. pittii* (35), *A. proteolyticus* (2), *A. radioresistens* (49), *A. schindleri* (3), *A. soli* (10), *A. towneri* (2), *A. ursingii* (83), *A. variabilis* (6), *A. vivianii* (1), unspeciated *Acinetobacter* (33)

Table 3 Activity of minocycline and comparator antimicrobial agents tested against 411 *Achromobacter* spp. isolates

Antimicrobial agent	No. of isolates	mg/L MIC_{50}	MIC_{90}	MIC range	CLSI[a] %S	%I	%R	EUCAST[a] %S	%I	%R
Minocycline	411	1	4	≤0.06 to >8	92.7	5.4	1.9			
Meropenem-vaborbactam	411	0.12	4	0.03 to >32						
Meropenem	411	0.25	16	0.03 to >32	85.9	3.9	10.2			
Imipenem	411	1	4	0.25 to >8	90.3	4.9	4.9			
Amikacin	411	>32	>32	≤0.25 to >32	9.5	5.6	84.9			
Aztreonam	411	>16	>16	2 to >16	1.0	1.0	98.1			
Cefepime	411	>16	>16	≤0.12 to >16	10.7	27.3	62.0			
Ceftazidime	411	4	16	0.25 to >16	76.9	13.1	10.0			
Gentamicin	411	>8	>8	≤0.12 to >8	6.1	2.7	91.2			
Levofloxacin	411	4	>4	≤0.12 to >4	36.0	30.7	33.3			
Piperacillin-tazobactam	411	≤0.5	16	≤0.5 to >64	92.2	3.6	4.1			

[a] Criteria as published in the "Other non-*Enterobacterales*" table in M100. (CLSI, 2020). EUCAST does not have breakpoints for this genus.
Organisms include: *Achromobacter denitrificans* (12), *A. insolitus* (6), *A. marplatensis* (1), *A. xylosoxidans* (202), unspeciated *Achromobacter* (190)

Figure 1 Infection types from which non-fermentative Gram-negative (NFGN) species were isolated, number of isolates

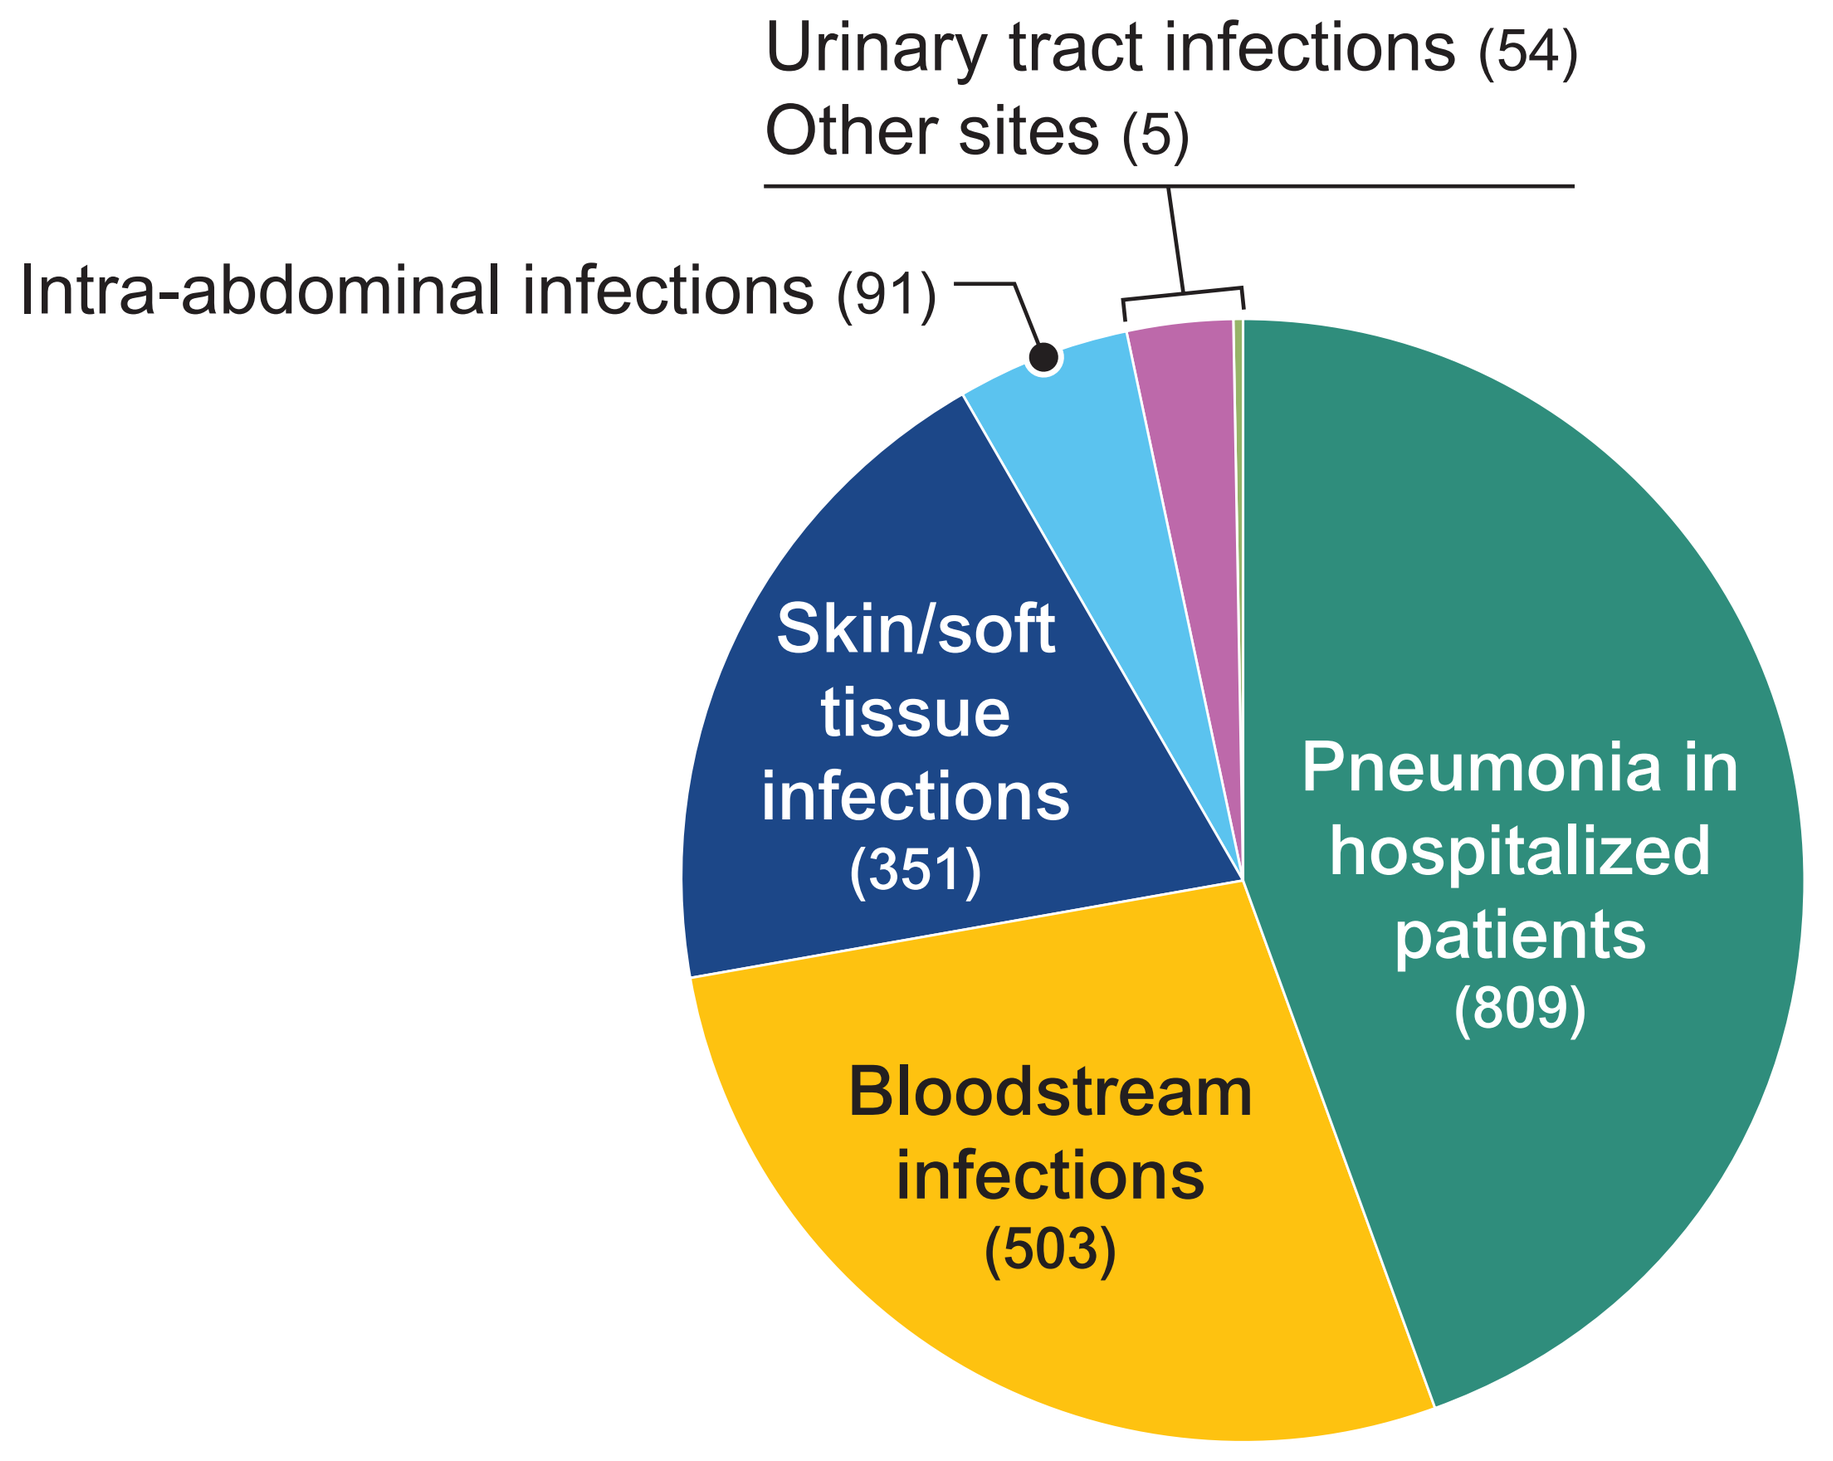
Urinary tract infections (54)
Other sites (5)
Intra-abdominal infections (91)
Skin/soft tissue infections (351)
Pneumonia in hospitalized patients (809)
Bloodstream infections (503)